Cyclophilin D knockout promotes cell death pathways in preventing HCC development in a streptozotocin-induced mouse model of diabetes-linked NASH

W. T. Stauffer[1], J. Kuo[2], M. Bobardt[1], D. R. Ure[3], R. T. Foster[3], and P. Gallay[1]

[1]Department of Immunology & Microbiology, Scripps Research, La Jolla, California 92037, USA.
[2]Arena Pharmaceuticals, San Diego, California 92121, USA.
[3]Hepion Pharmaceuticals, Edison, New Jersey 08837, USA.

EASL The Home of Hepatology

Scripps Research
Science Changing Life

PREMISE

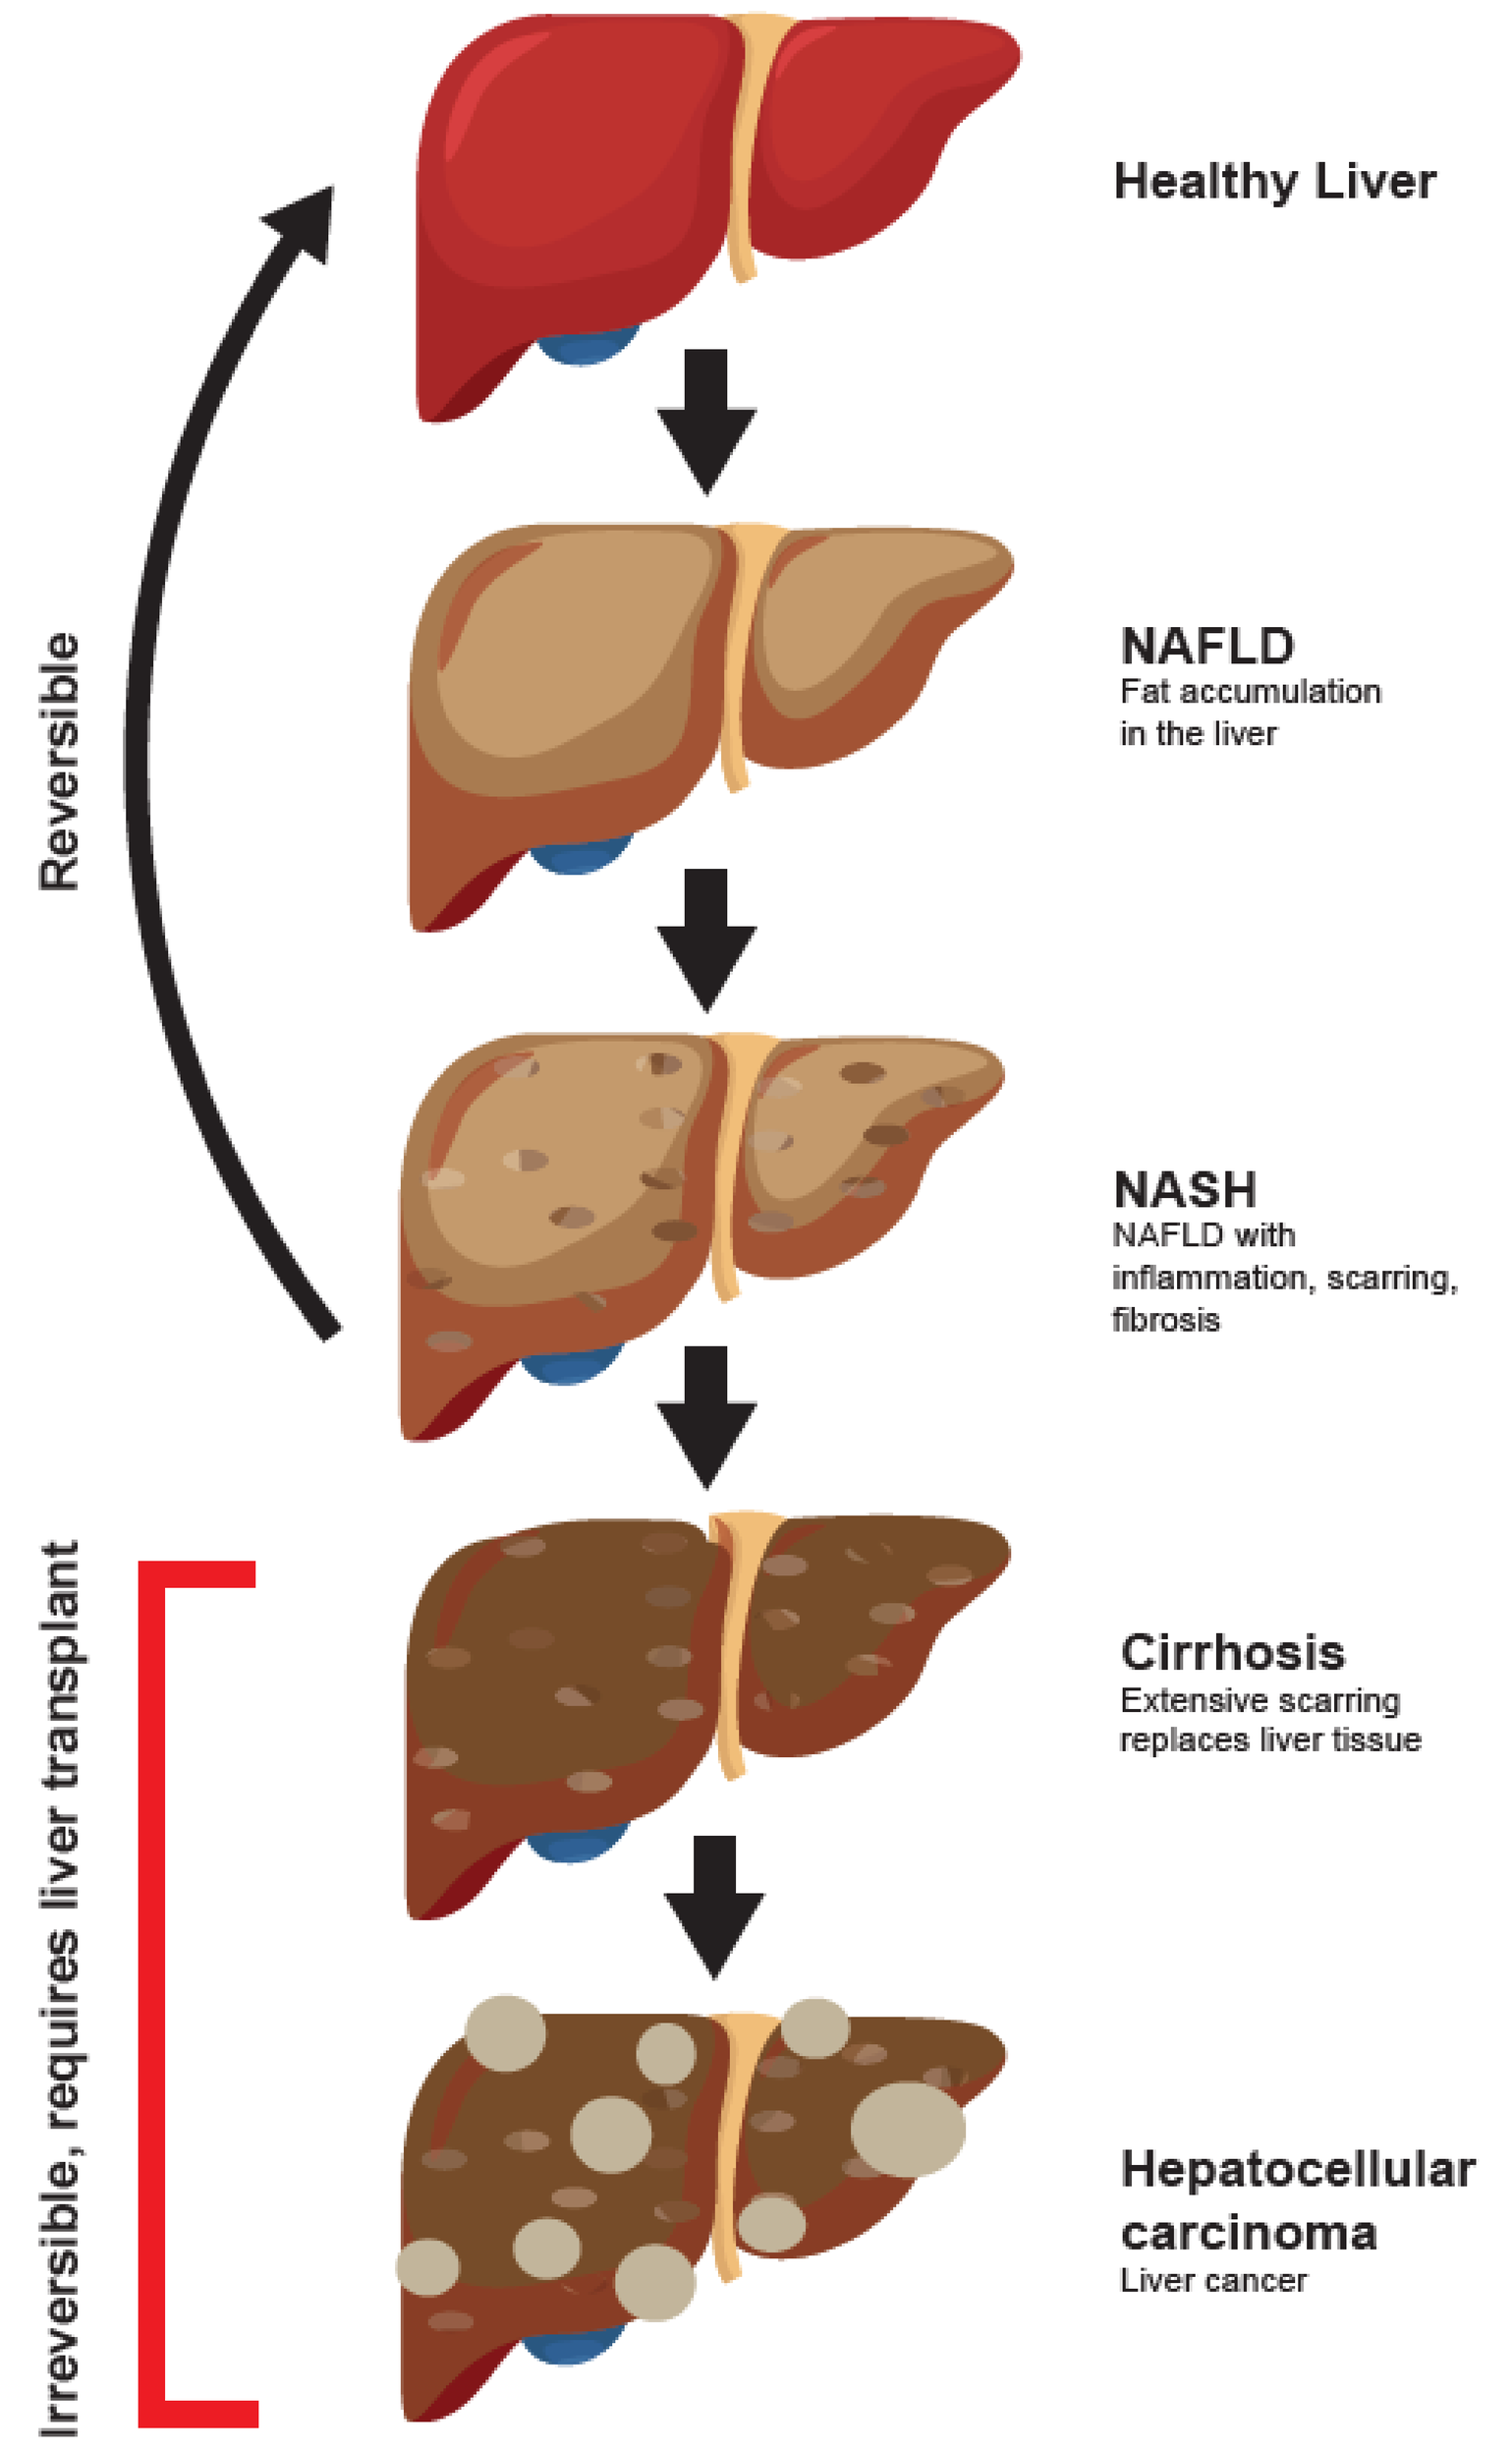
Healthy Liver
NAFLD
Fat accumulation in the liver
NASH
NAFLD with inflammation, scarring, fibrosis
Cirrhosis
Extensive scarring replaces liver tissue
Hepatocellular carcinoma
Liver cancer
Reversible
Irreversible, requires liver transplant

- ~25% of the adult population has some form of fatty liver.
- Associated with obesity, diabetes, and metabolic syndrome.
- Non-alcoholic fatty liver disease (NAFLD) features simple steatosis or lipid accumulation.
- NAFLD may progress to non-alcoholic steato-hepatitis (NASH) with inflammation and hepatocyte damage.
- NAFLD and NASH can be reversed.
- Further scarring can induce cirrhosis and the development of hepatocellular carcinoma (HCC), which cannot be reversed.

- Previous studies[1,2] have shown cyclophilin (Cyp) inhibitor drugs can prevent HCC development. It is unknown which Cyp family member(s) is the cause.
- Cyclophilins are a family of peptidylprolyl isomerases with diverse functions. CypD is a component of the mitochondrial permeability transition pore (mPTP).

METHODS

C57BL/6J WT Pregnant Dams | C57BL/6J *Ppif -/-* Pregnant Dams

Birth of pups, select for males | Birth of pups, select for males

+ 200µg streptozotocin (STZ, Sigma-Aldrich) per pup on day P2 (n=10 per group)

wean at three weeks old

Start *ad libitum* "western diet" with 21.1% fat, 41% sucrose, and 1.25% cholesterol by weight and high sugar solution (23.1g/L fructose and 18.9g/L glucose)

Continue western diet for 27 weeks | **Development of NAFLD, NASH, and HCC** | Continue western diet for 27 weeks

Sacrifice WT and *Ppif -/-* mice at 30-weeks old for HCC timepoint

- The STAM model[3] mimics diabetes in mice by destroying pancreatic beta cells.
- Combined with western diet, STAM reliably produces NAFLD/NASH and HCC.
- Comparing wild-type (WT) mice to *Ppif* (CypD) knockout (KO) mice reveals the necessity for CypD in HCC development.

RESULTS

WT and CypD KO Mice Both Develop NASH in the STAM Model

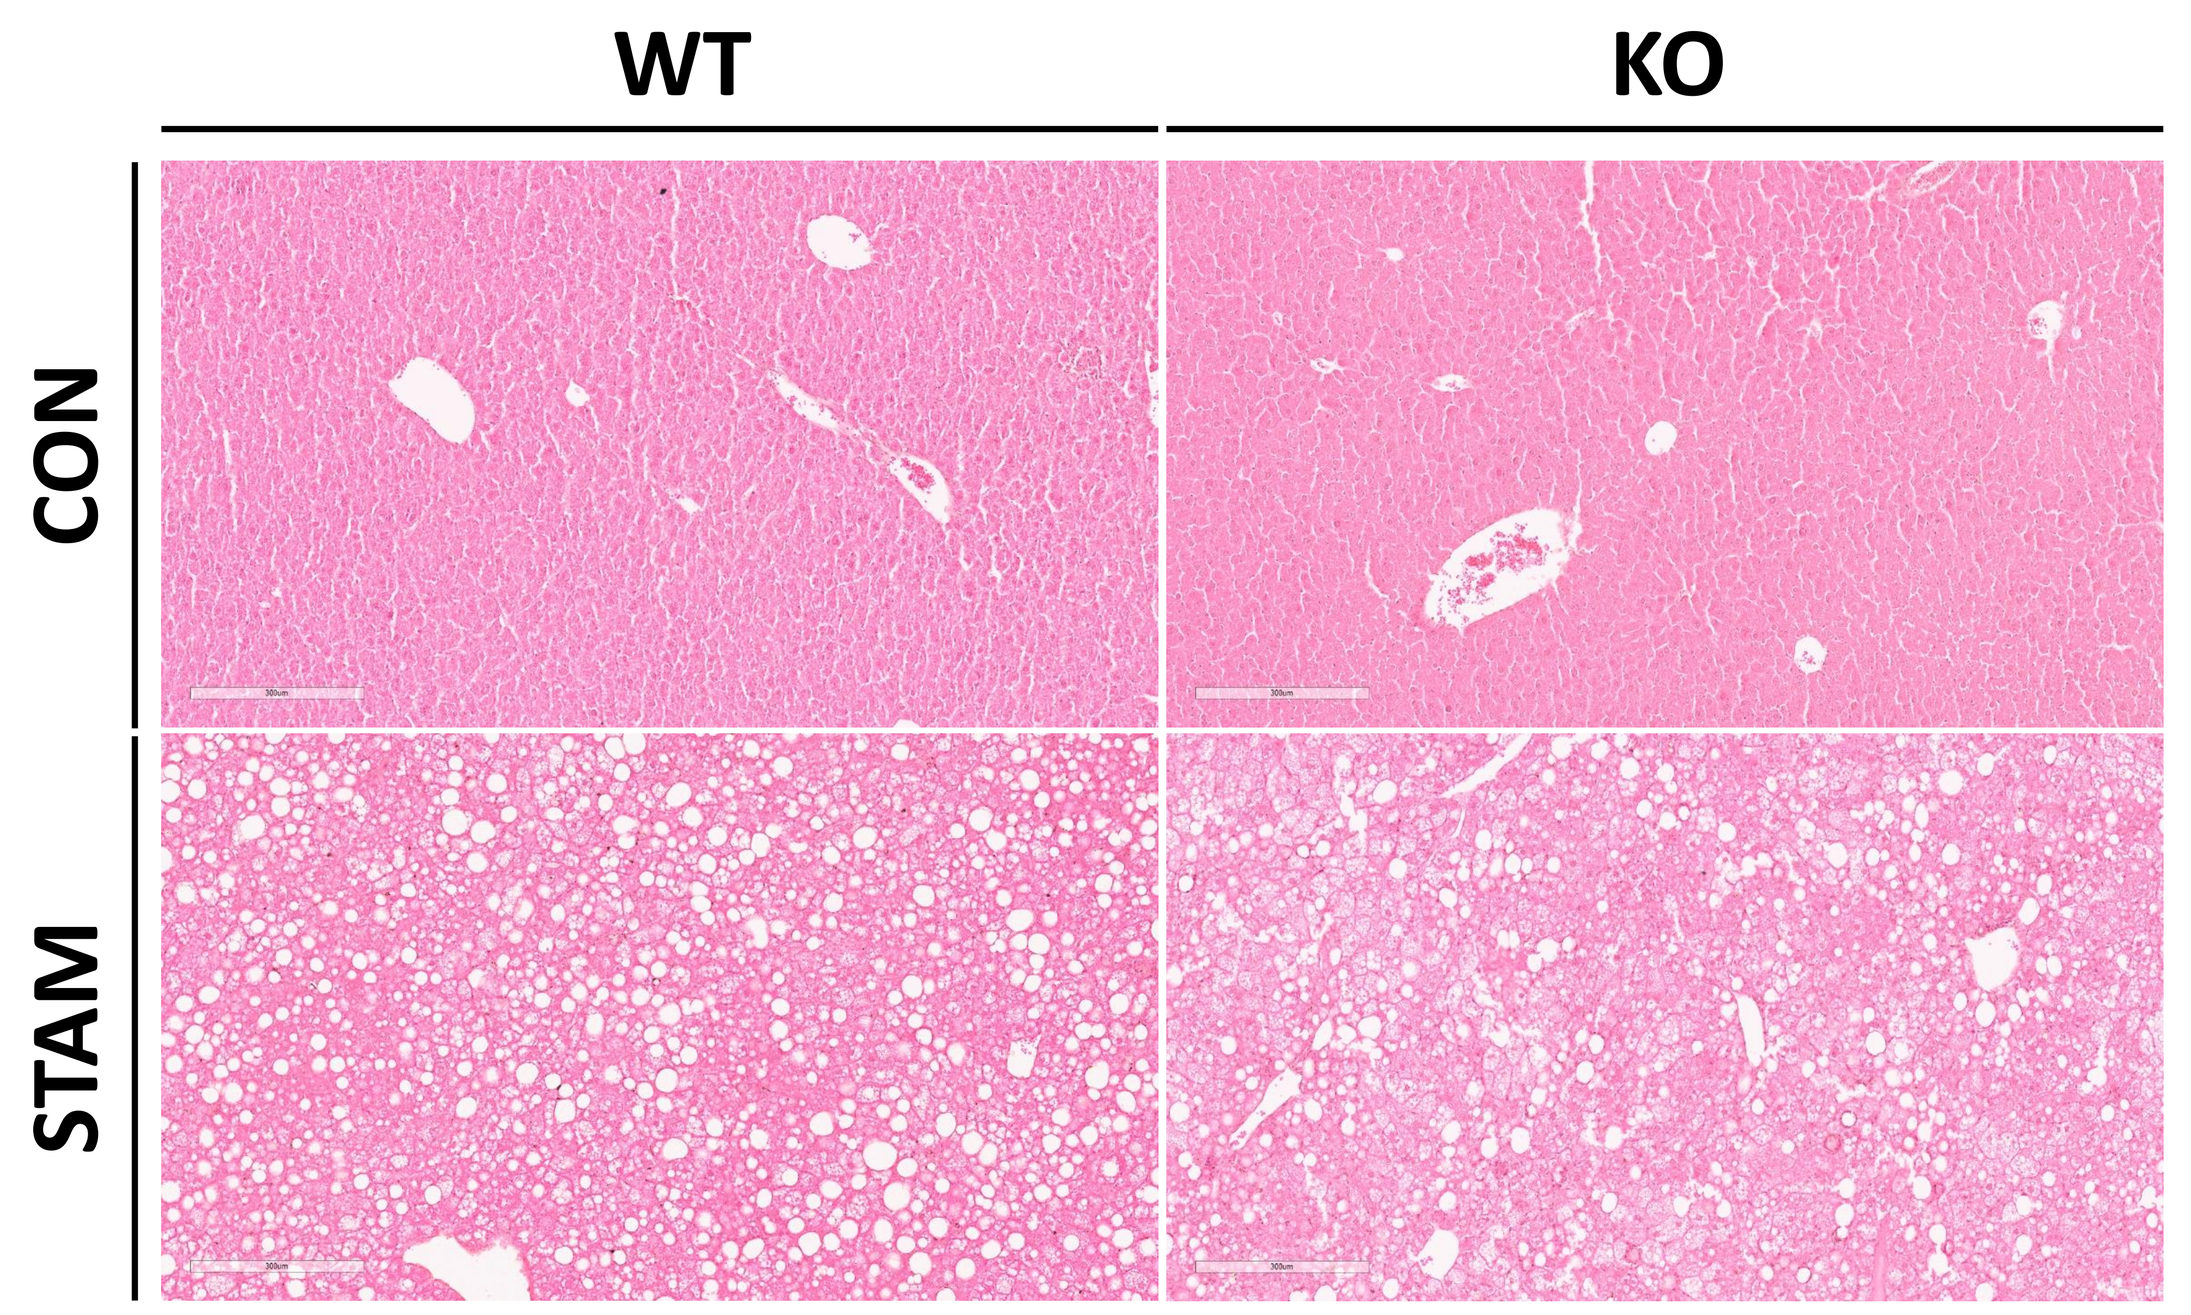
WT
KO
CON
STAM

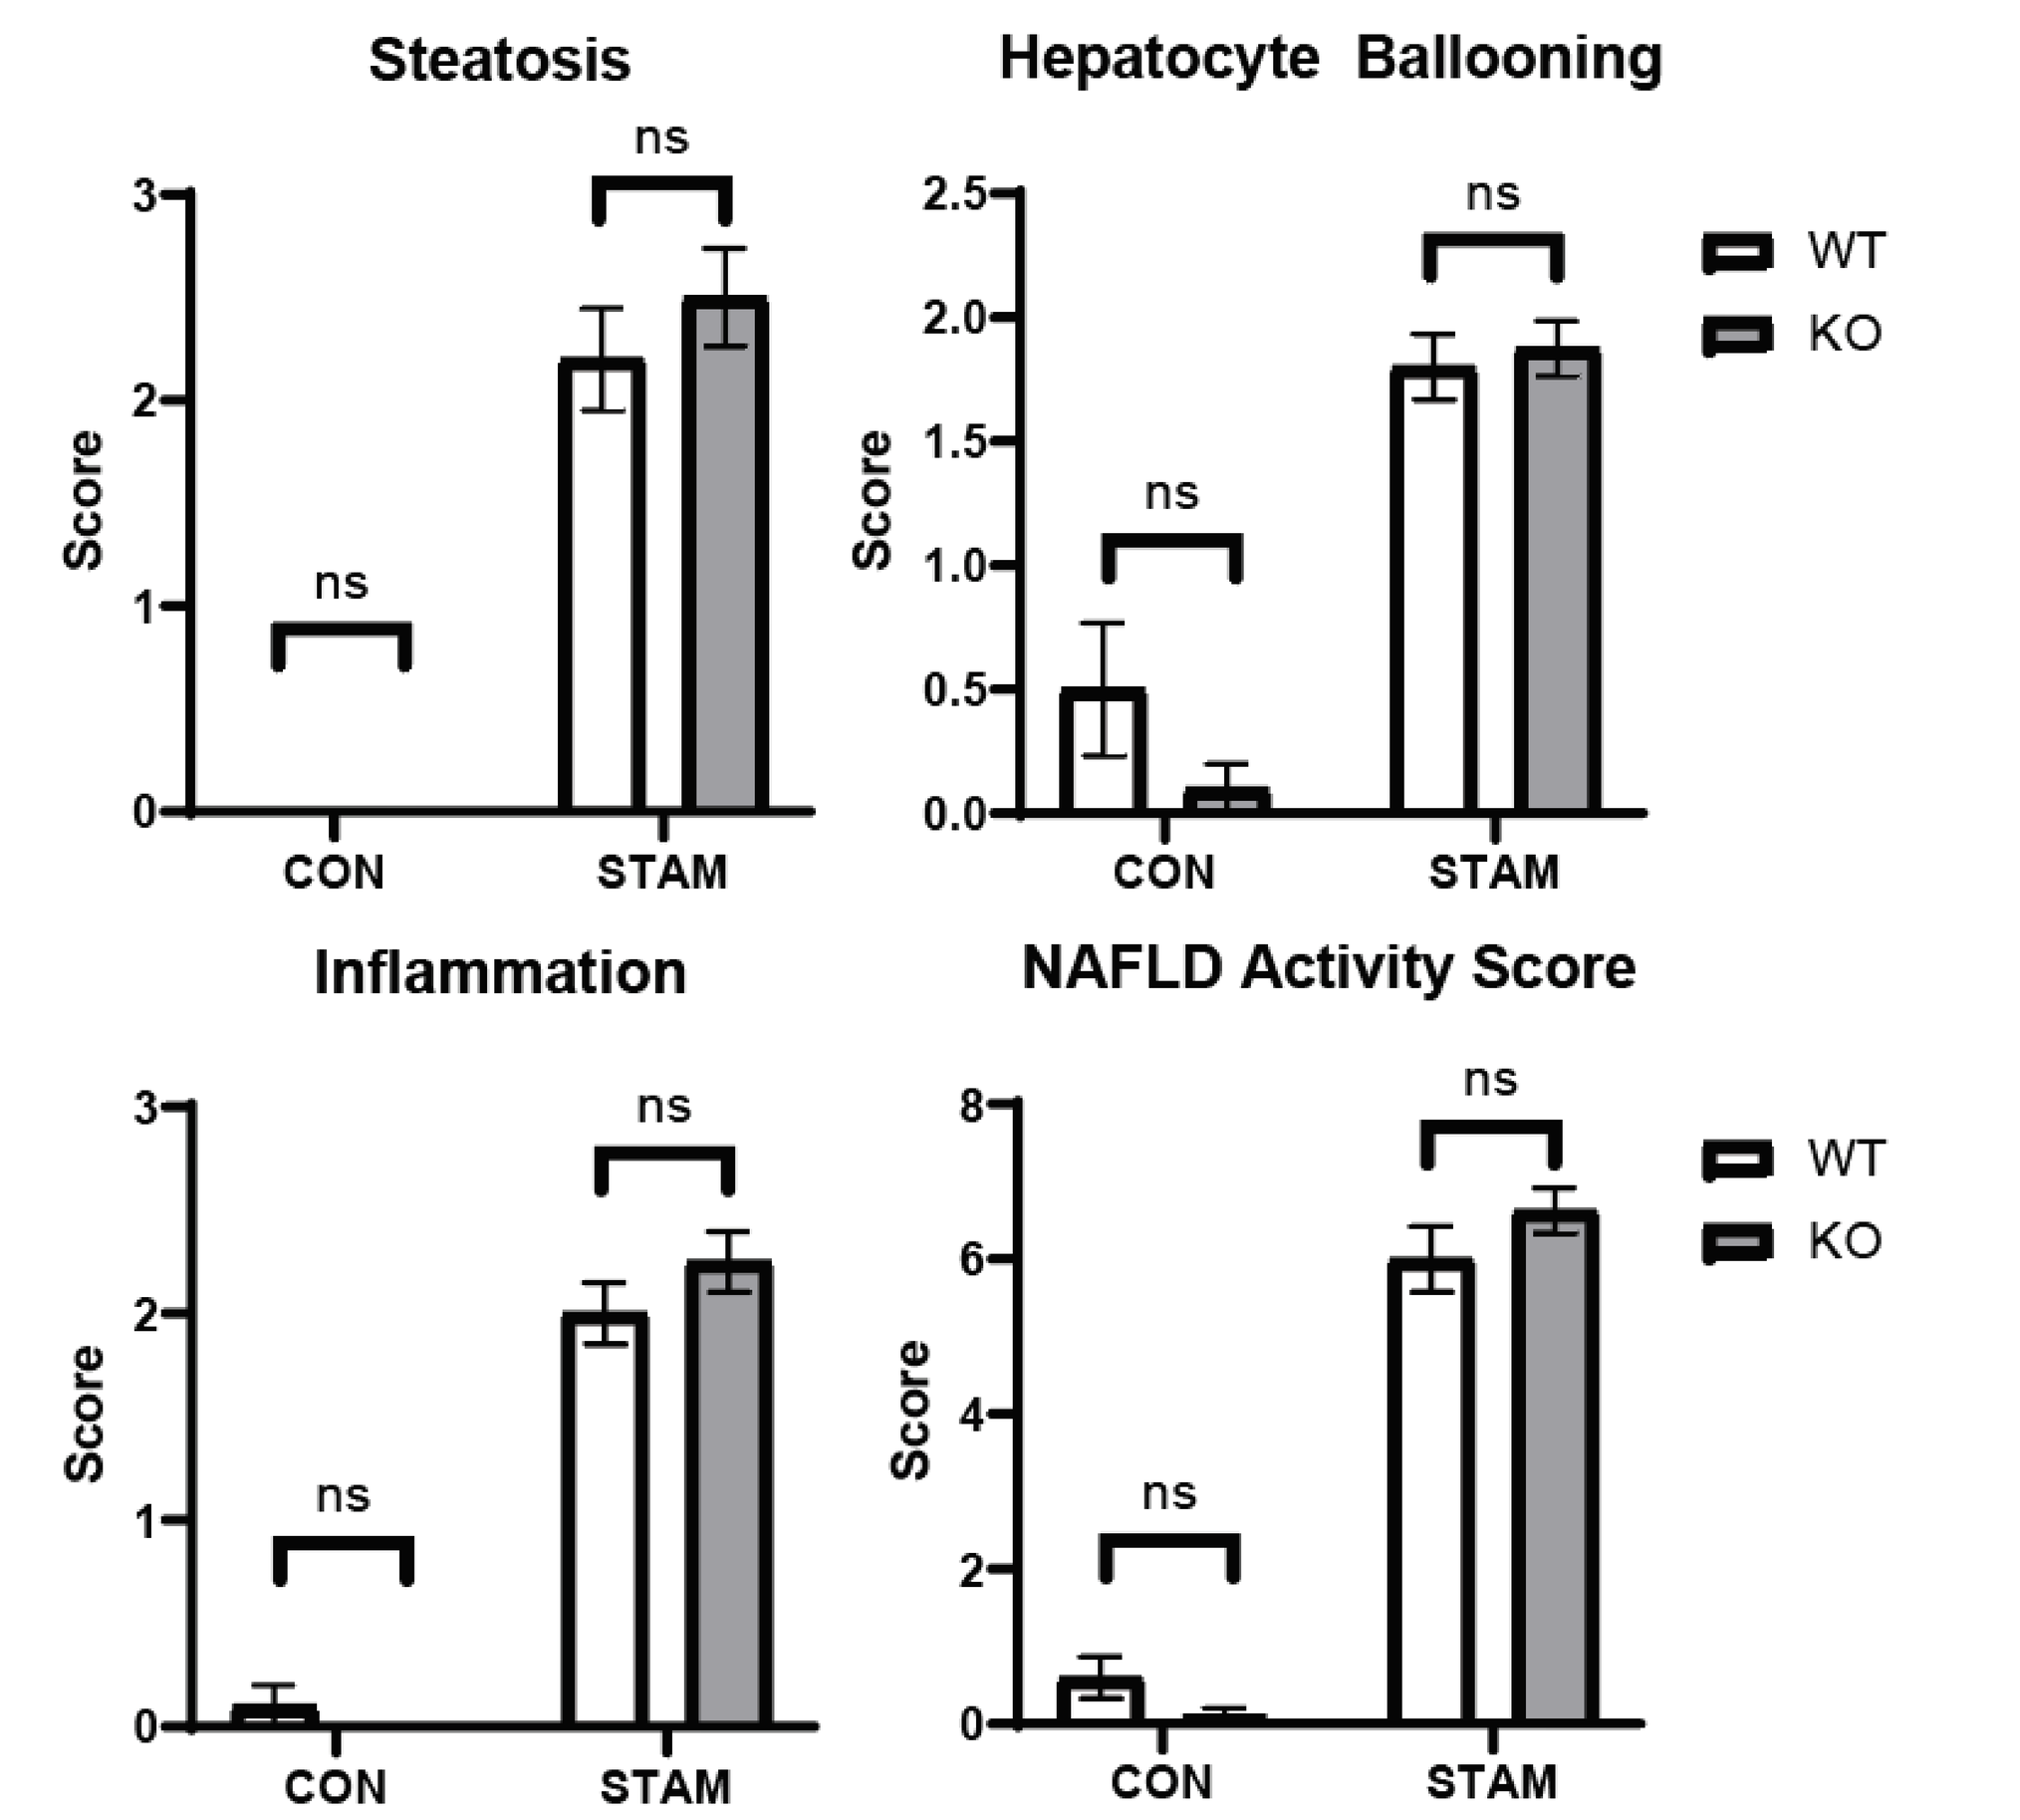
Steatosis
Hepatocyte Ballooning
Inflammation
NAFLD Activity Score
Score
ns
CON
STAM
WT
KO

- Under STAM, both WT and CypD KO mice developed significant steatosis, hepatocyte ballooning, and inflammation, as determined by histological analysis of hematoxylin & eosin (H&E)-stained liver tissue sections.
- Combining scores for all three categories yields the NAFLD activity score (NAS). Both WT and KO STAM mice had average NAS of greater than 6, which is consistent with NASH. ns=not significant (p>0.05) by unpaired student's t-test. Representative images of H&E-stained liver sections are above on the left.

- Our scoring system for NAS is as follows: for ballooning: 0 (no changes), 1 (few ballooned cells), and 2 (many prominent balloon cells); for inflammation: 0 (no changes), 1 (minimal infiltration with no major inflammatory foci), 2 (mild with <2 inflammatory clusters), 3 (moderate with 2–4 foci of leukocytes), and 4 (severe infiltration with >4 inflammatory clusters); for steatosis: 0 (<5% steatosis), 1 (5–33%), 2 (33–66%), and 3 (>66% steatosis).

CypD Knockout Prevents HCC Development in the STAM Model

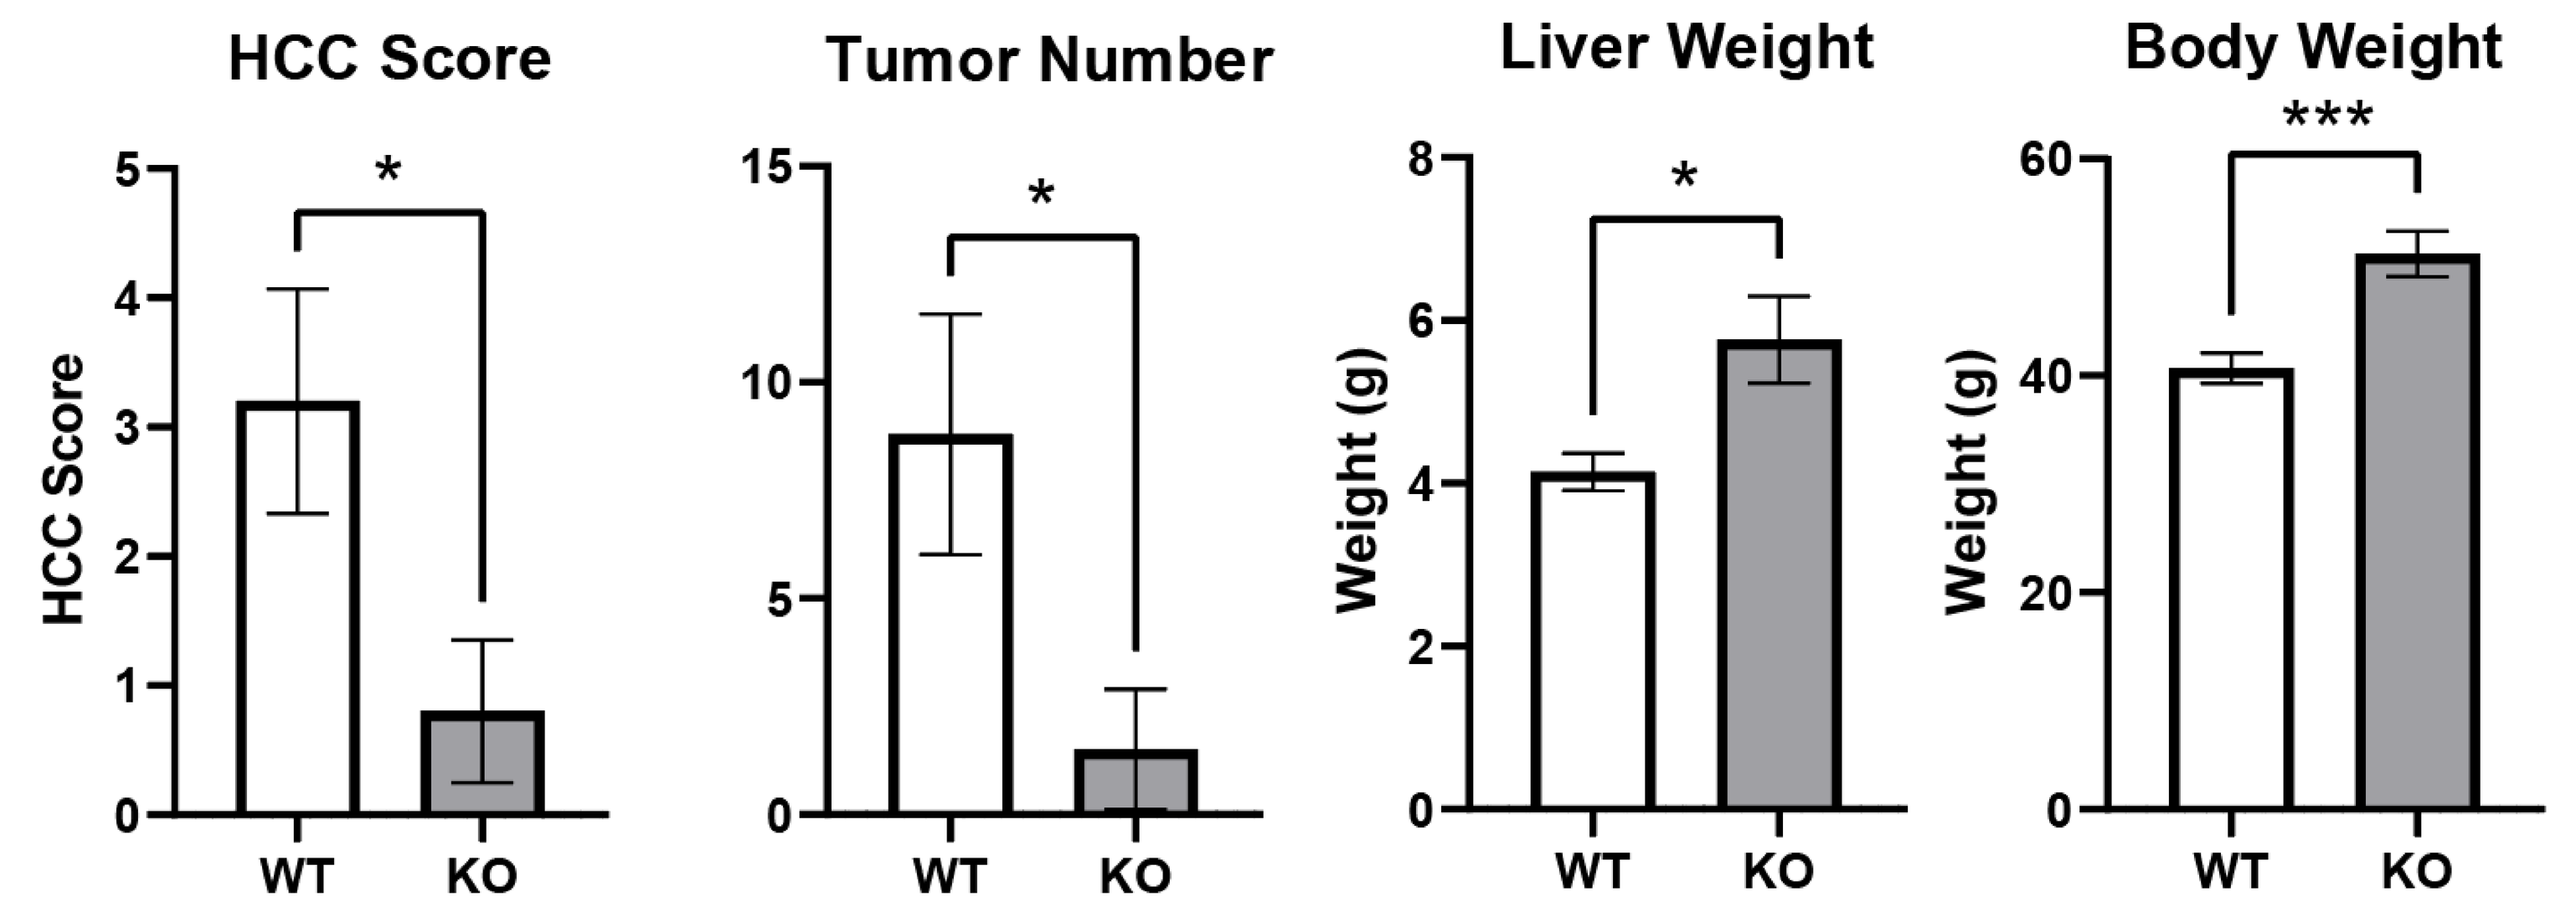
HCC Score
Tumor Number
Liver Weight
Body Weight
HCC Score
Weight (g)
*

WT
KO

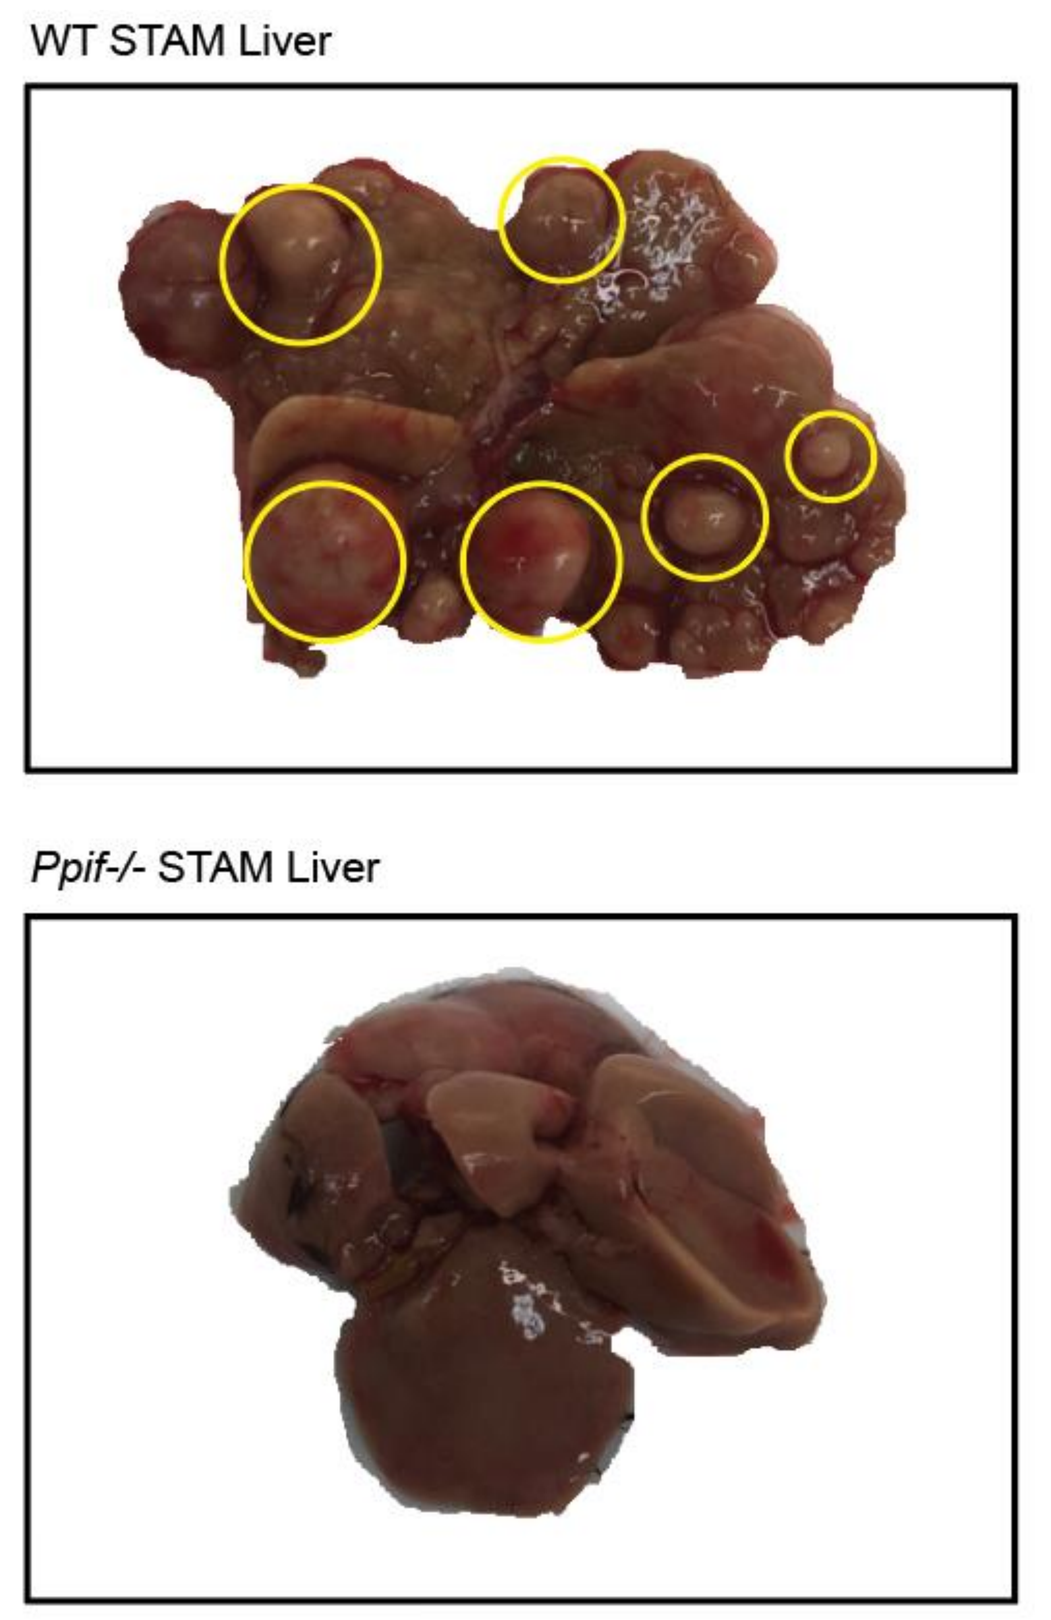
WT STAM Liver
Ppif-/- STAM Liver

- CypD KO mice developed significantly less tumor burden after STAM compared to WT. *p≤0.05, ***p≤0.001 by unpaired student's t-test. Representative images of livers are on the right. HCC tumors are circled.
- Our scoring system for tumor burden is as follows: Scores included 0 (no detectable nodules), 1 (no more than 4 small ≤0.5cm nodules with no larger nodules), 2 (more than 4 small nodules with no larger nodules), 3 (no more than 2 medium-sized (<1cm) nodules with no larger nodules), 4 (3 or more medium-sized nodules with no larger nodules), 5 (no more than 1 large (≥1cm) nodule), 6 (no more than 2 large nodules), and 7 (3 or more large nodules).

CypD KO in the STAM Model Suppresses the Oncogene *Birc5*

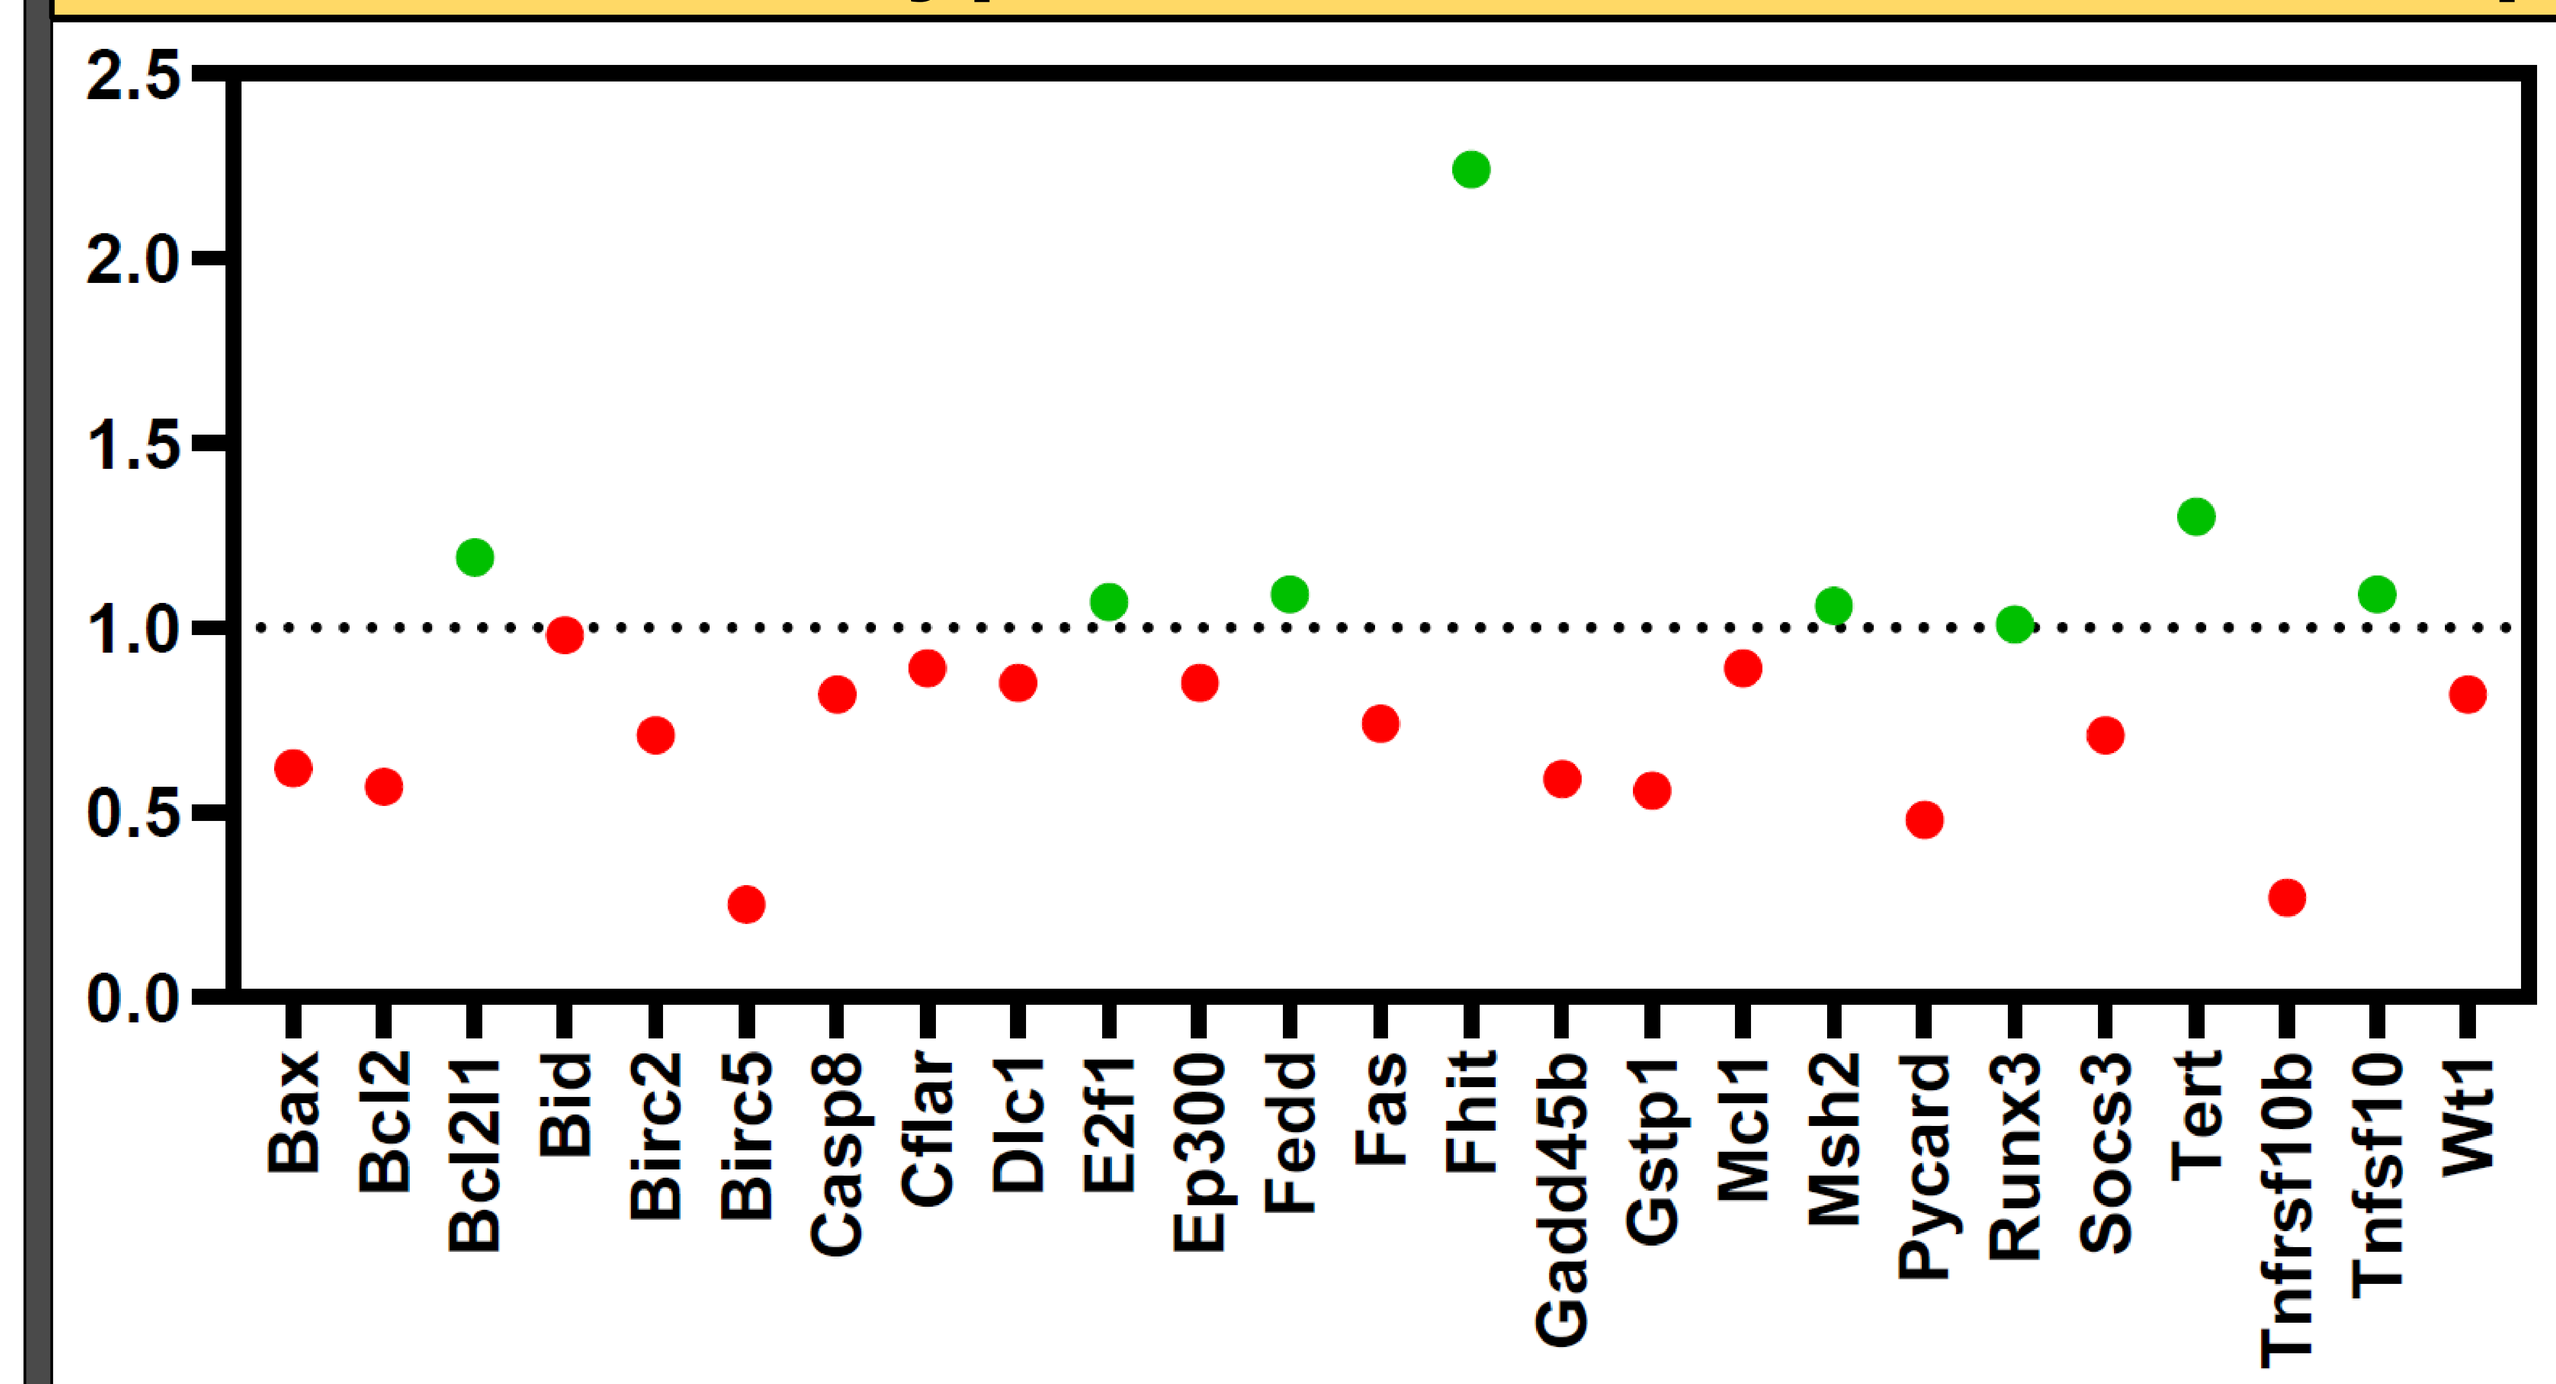
Bax
Bcl2
Bcl2l1
Bid
Birc2
Birc5
Casp8
Cflar
Dlc1
E2f1
Ep300
Fedd
Fas
Fhit
Gadd45b
Gstp1
Mcl1
Msh2
Pycard
Runx3
Socs3
Tert
Tnfrsf10b
Tnfsf10
Wt1

- To explore possible consequences of CypD KO and impaired mPTP activity in HCC development, we conducted a preliminary PCR array on post-STAM WT and CypD KO livers of relevant apoptosis and cell death gene transcripts.
- While a majority of apoptotic gene-transcripts were down regulated in CypD KO STAM livers relative to WT (red dots), some anti-apoptotic genes-transcripts were also decreased, most especially *Birc5* (red dots).
- *Birc5* is anti-apoptotic and is elevated in a variety of cancers, including HCC, though it is not known whether it is regulated by CypD.
- Further investigation is required to confirm these findings and determine if there is a mechanistic link between *Birc5* induction and HCC prevention in this model.

CONCLUSIONS

- CypD KO prevents in the development of HCC in this model of diabetes-induced NAFLD/NASH.
- This suggests CypD inhibition is a viable target for the treatment of NAFLD/NASH and is in agreement with previous studies which showed global cyclophilin-inhibitor drugs had similar effects.
- Future studies include determining the mechanism(s) of action of CypD KO in preventing HCC development. Furthermore, it must be determined whether the role of CypD in the mPTP or the suppression of Birc5 play roles critical to this process.

REFERENCES

1. Kuo J et al. A Pan-Cyclophilin Inhibitor, CRV431, Decreases Fibrosis and Tumor Development in Chronic Liver Disease Models. *J Pharmacol Exp Ther.* 2019; 371(2):231-241.
2. Kuo J et al. Cyclophilin Inhibitor NV556 Reduces Fibrosis and Hepatocellular Carcinoma Development in Mice With Non-Alcoholic Steatohepatitis. *Front Pharmacol.* 2019; 10:1129.
3. Fujii et al. A murine model for non-alcoholic steatohepatitis showing evidence of association between diabetes and hepatocellular carcinoma. *Med Mol Morphol.* 2013; 46:141-152.

DISCLOSURES

- This project was funded by the National Institute of Allergy and Infectious Diseases of the National Institutes of Health under award number R01AI143931-02 (Gallay).
- Stauffer was supported by the Molecular Basis of Viral Pathogenesis Postdoctoral Training Grant (T32AI007354).
- Stauffer, Kuo, Bobardt, and Gallay declare no competing financial interests. Ure and Foster are employees of Hepion Pharmaceuticals Inc.

PosterSessionOnline.com
Scan to download the poster
THE INTERNATIONAL LIVER CONGRESS™
DOI: 10.3252/pso.eu.ILC2022.2022
Liver tumors
Winston Stauffer
FRI--568
ILC2022