Rencofilstat, a pan-cyclophilin inhibitor, exerts diverse metabolic and transcriptional anti-tumor activities in a murine NASH-HCC model

D. URE[1], J. KUO[2], W. STAUFFER[2], L. HADDON[1], C. FU[1], P. MAYO[1], R. FOSTER[1], P. GALLAY[2]
[1] Hepion Pharmaceuticals, Edmonton, Canada, and Edison, USA [2] Scripps Research Institute, La Jolla, USA

INTRODUCTION

Cyclophilins are protein folding and regulatory enzymes that have been shown in preclinical studies to contribute to progression of several types of cancer, including hepatocellular carcinoma (HCC). They are elevated in most types of cancer, and overexpression is prognostic of shorter survival. Therefore, cyclophilin inhibition is proposed to have anti-cancer activities. Rencofilstat (CRV431) is a second-generation, clinical phase cyclophilin inhibitor being investigated in NASH and HCC.

AIM

To investigate the cyclophilin inhibitor, rencofilstat (CRV431) in a murine NASH-HCC model and conduct multi-omic analyses to identify mechanistic pathways through which rencofilstat affected HCC.

METHODS

Streptozotocin-Western diet Model of NASH-HCC (STAM™)

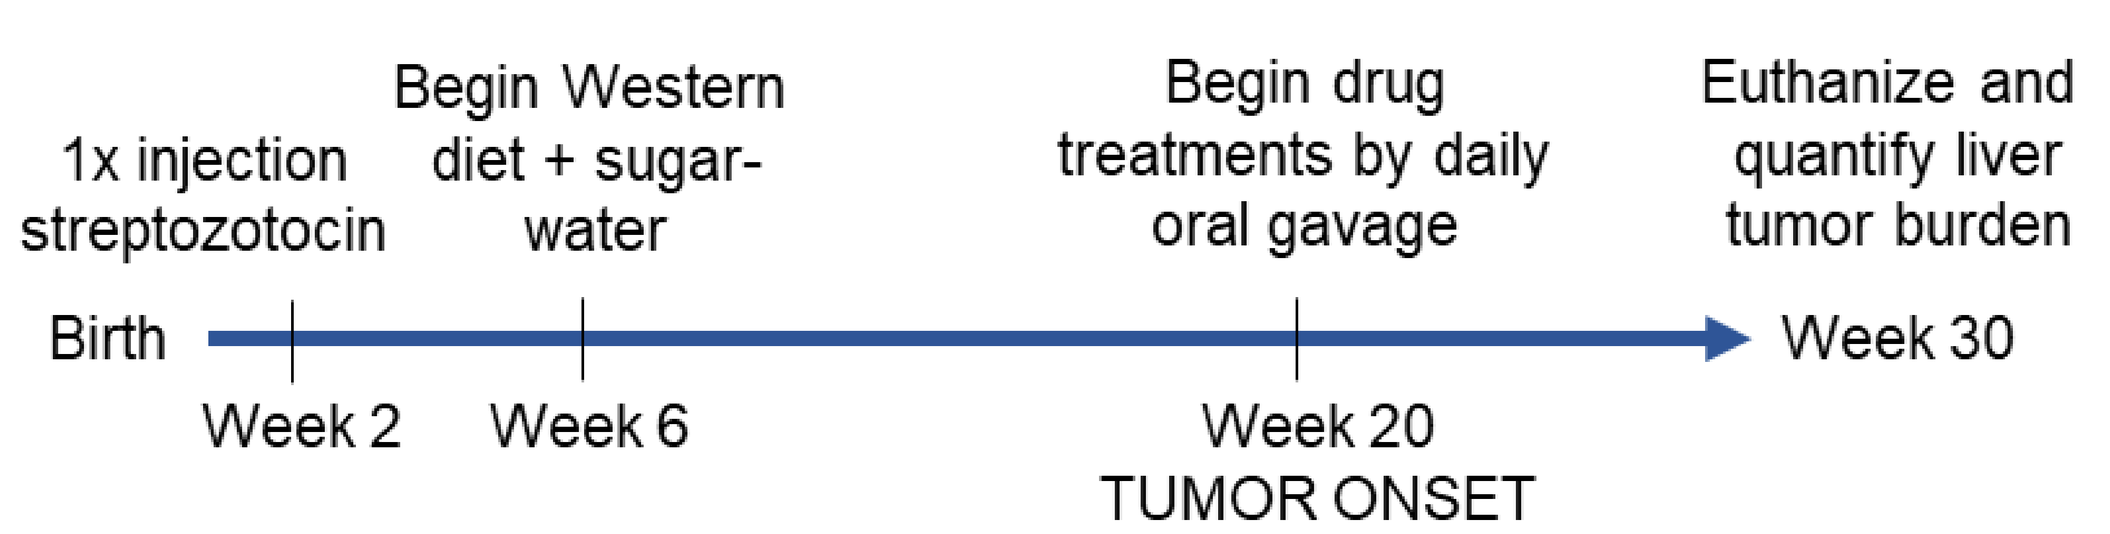
1x injection streptozotocin
Begin Western diet + sugar-water
Begin drug treatments by daily oral gavage
Euthanize and quantify liver tumor burden
Birth
Week 2
Week 6
Week 20
TUMOR ONSET
Week 30

Drug Treatments

Daily oral gavage of Vehicle or 3 different cyclophilin inhibitors from Weeks 20-30 of age

- Rencofilstat – clinical phase nonimmunosuppressive cyclosporin
- Debio-025 – 1st generation nonimmunosuppressive cyclosporin
- NV556 – nonclinical sangamide

Multi-Omics Analyses of Tumor and NASH Liver Tissue

- Whole exome sequencing: 36 samples
- RNA sequencing: 47 samples
- Untargeted metabolomics: 36 samples

CONTACT INFORMATION

Daren Ure, PhD
Chief Scientific Officer
Hepion Pharmaceuticals
dure@hepionpharma.com

RESULTS

Cyclophilin Inhibitors Decrease Tumor Burden

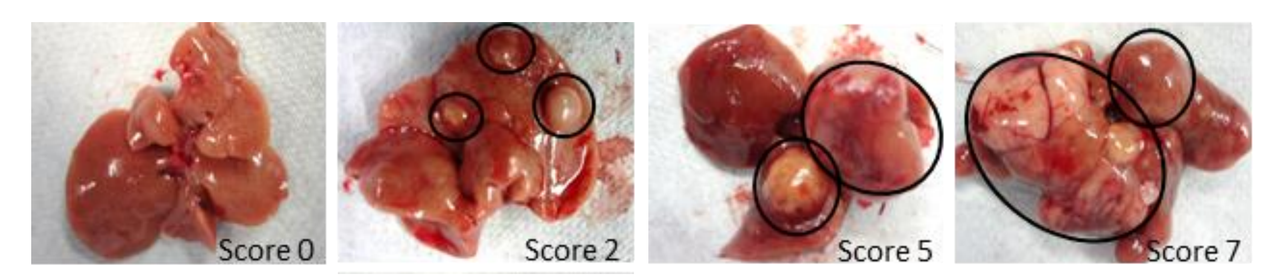
Score 0
Score 2
Score 5
Score 7

Fewer liver tumors developed in mice treated with cyclophilin inhibitors.

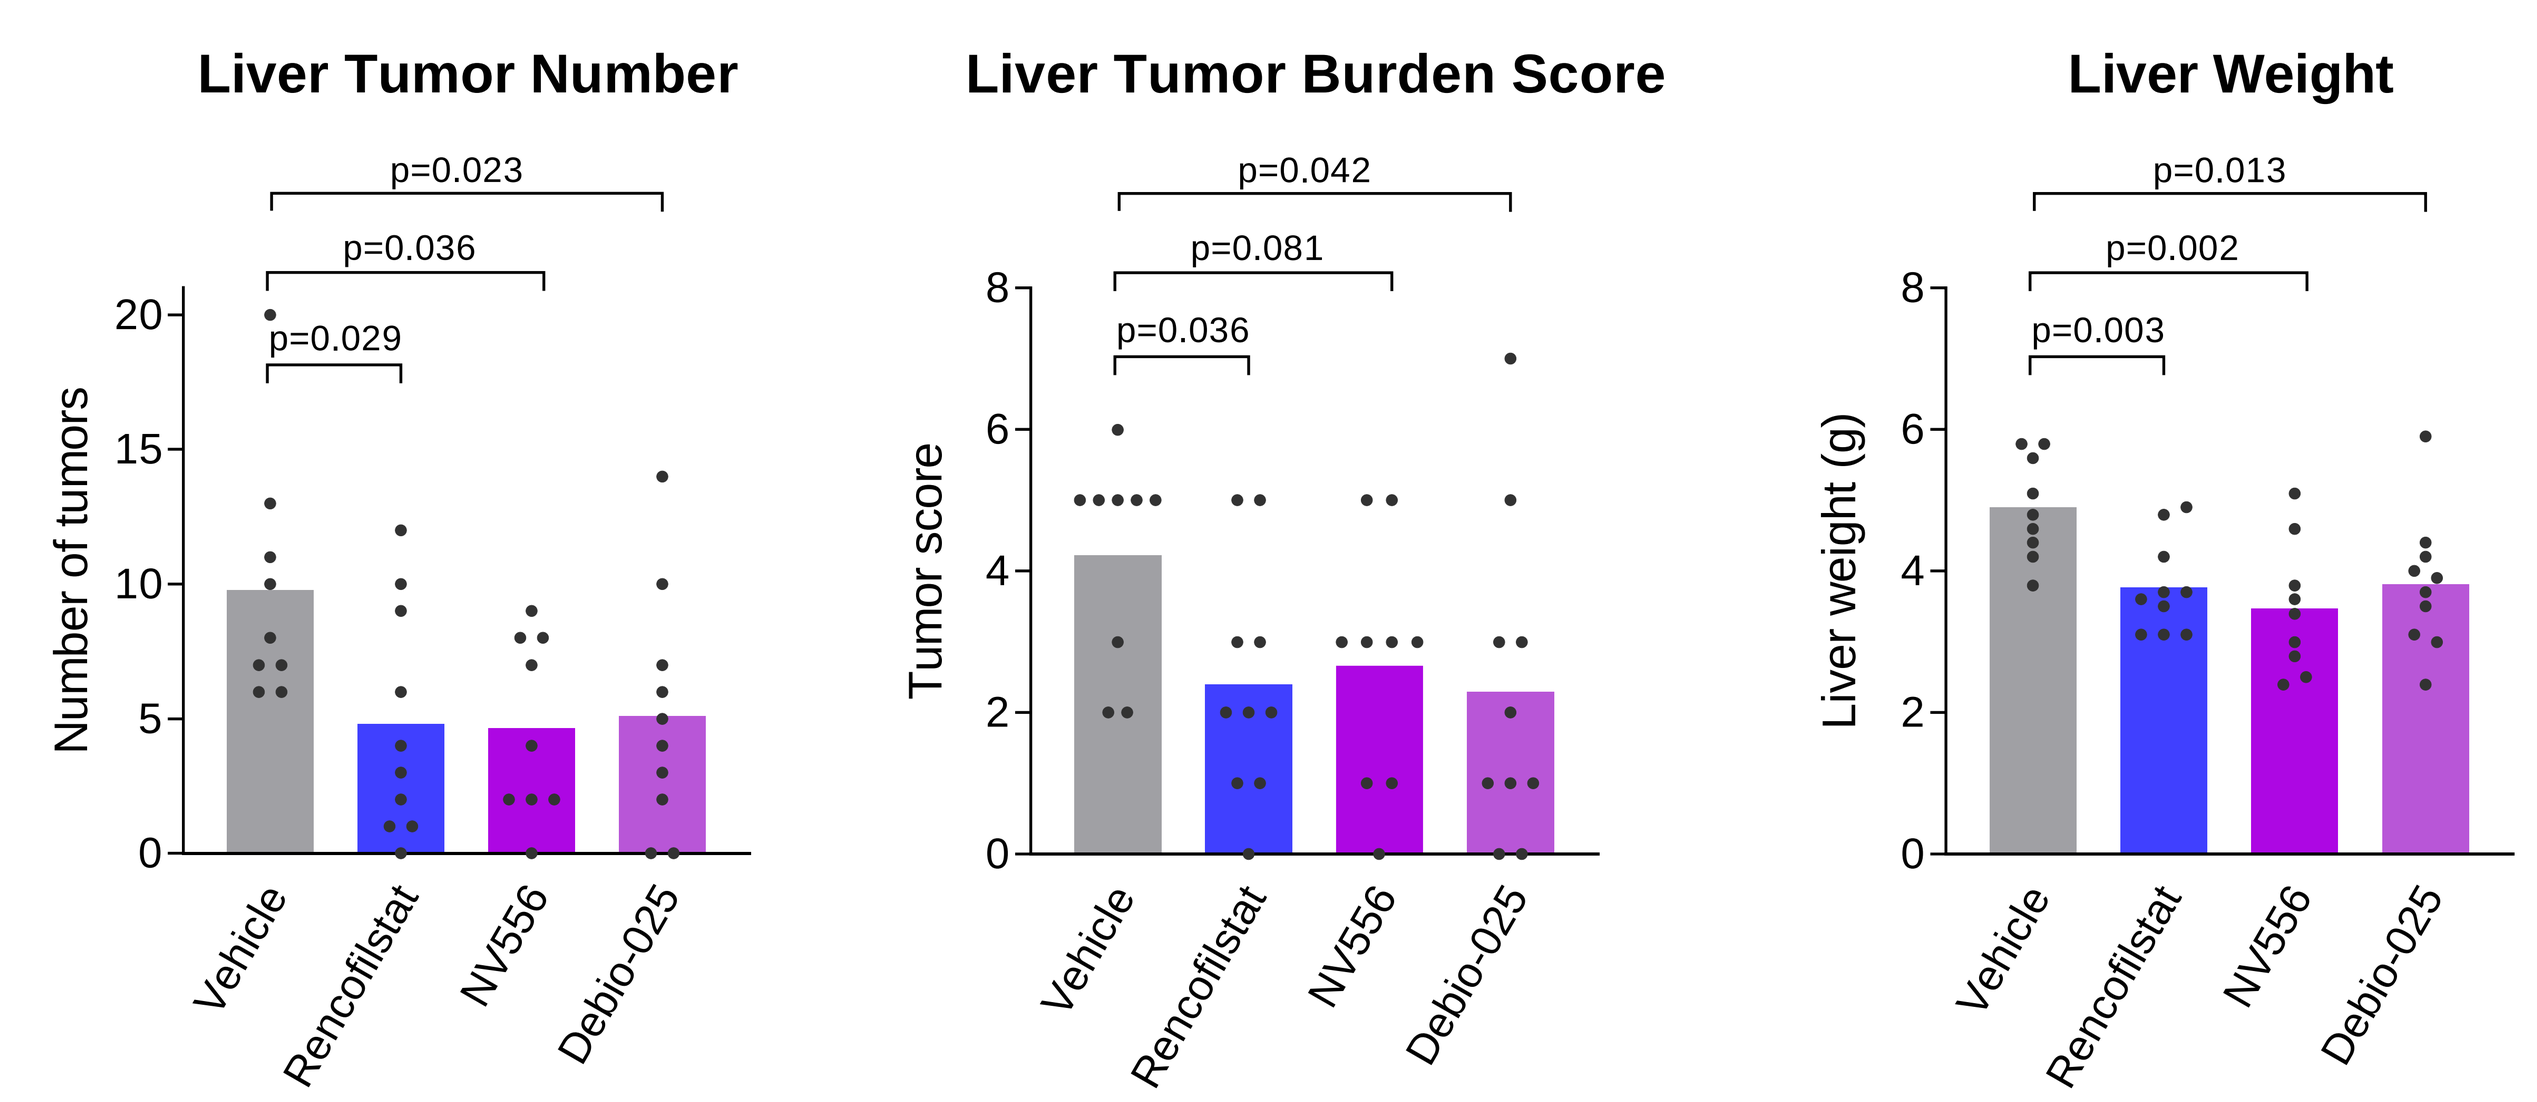
Liver Tumor Number
Liver Tumor Burden Score
Liver Weight
Number of tumors
Tumor score
Liver weight (g)
p=0.023
p=0.036
p=0.029
p=0.042
p=0.081
p=0.036
p=0.013
p=0.002
p=0.003
Vehicle
Rencofilstat
NV556
Debio-025

Mutation Analysis of Tumors and NASH Tissue

Tumors from vehicle-treated and rencofilstat-treated mice contained similarly high numbers of nonsynonymous DNA mutations in a diversity of human cancer driver genes. Mean tumor masses of analyzed tumors were 64 mg (Vehicle) and 65 mg (Rencofilstat).

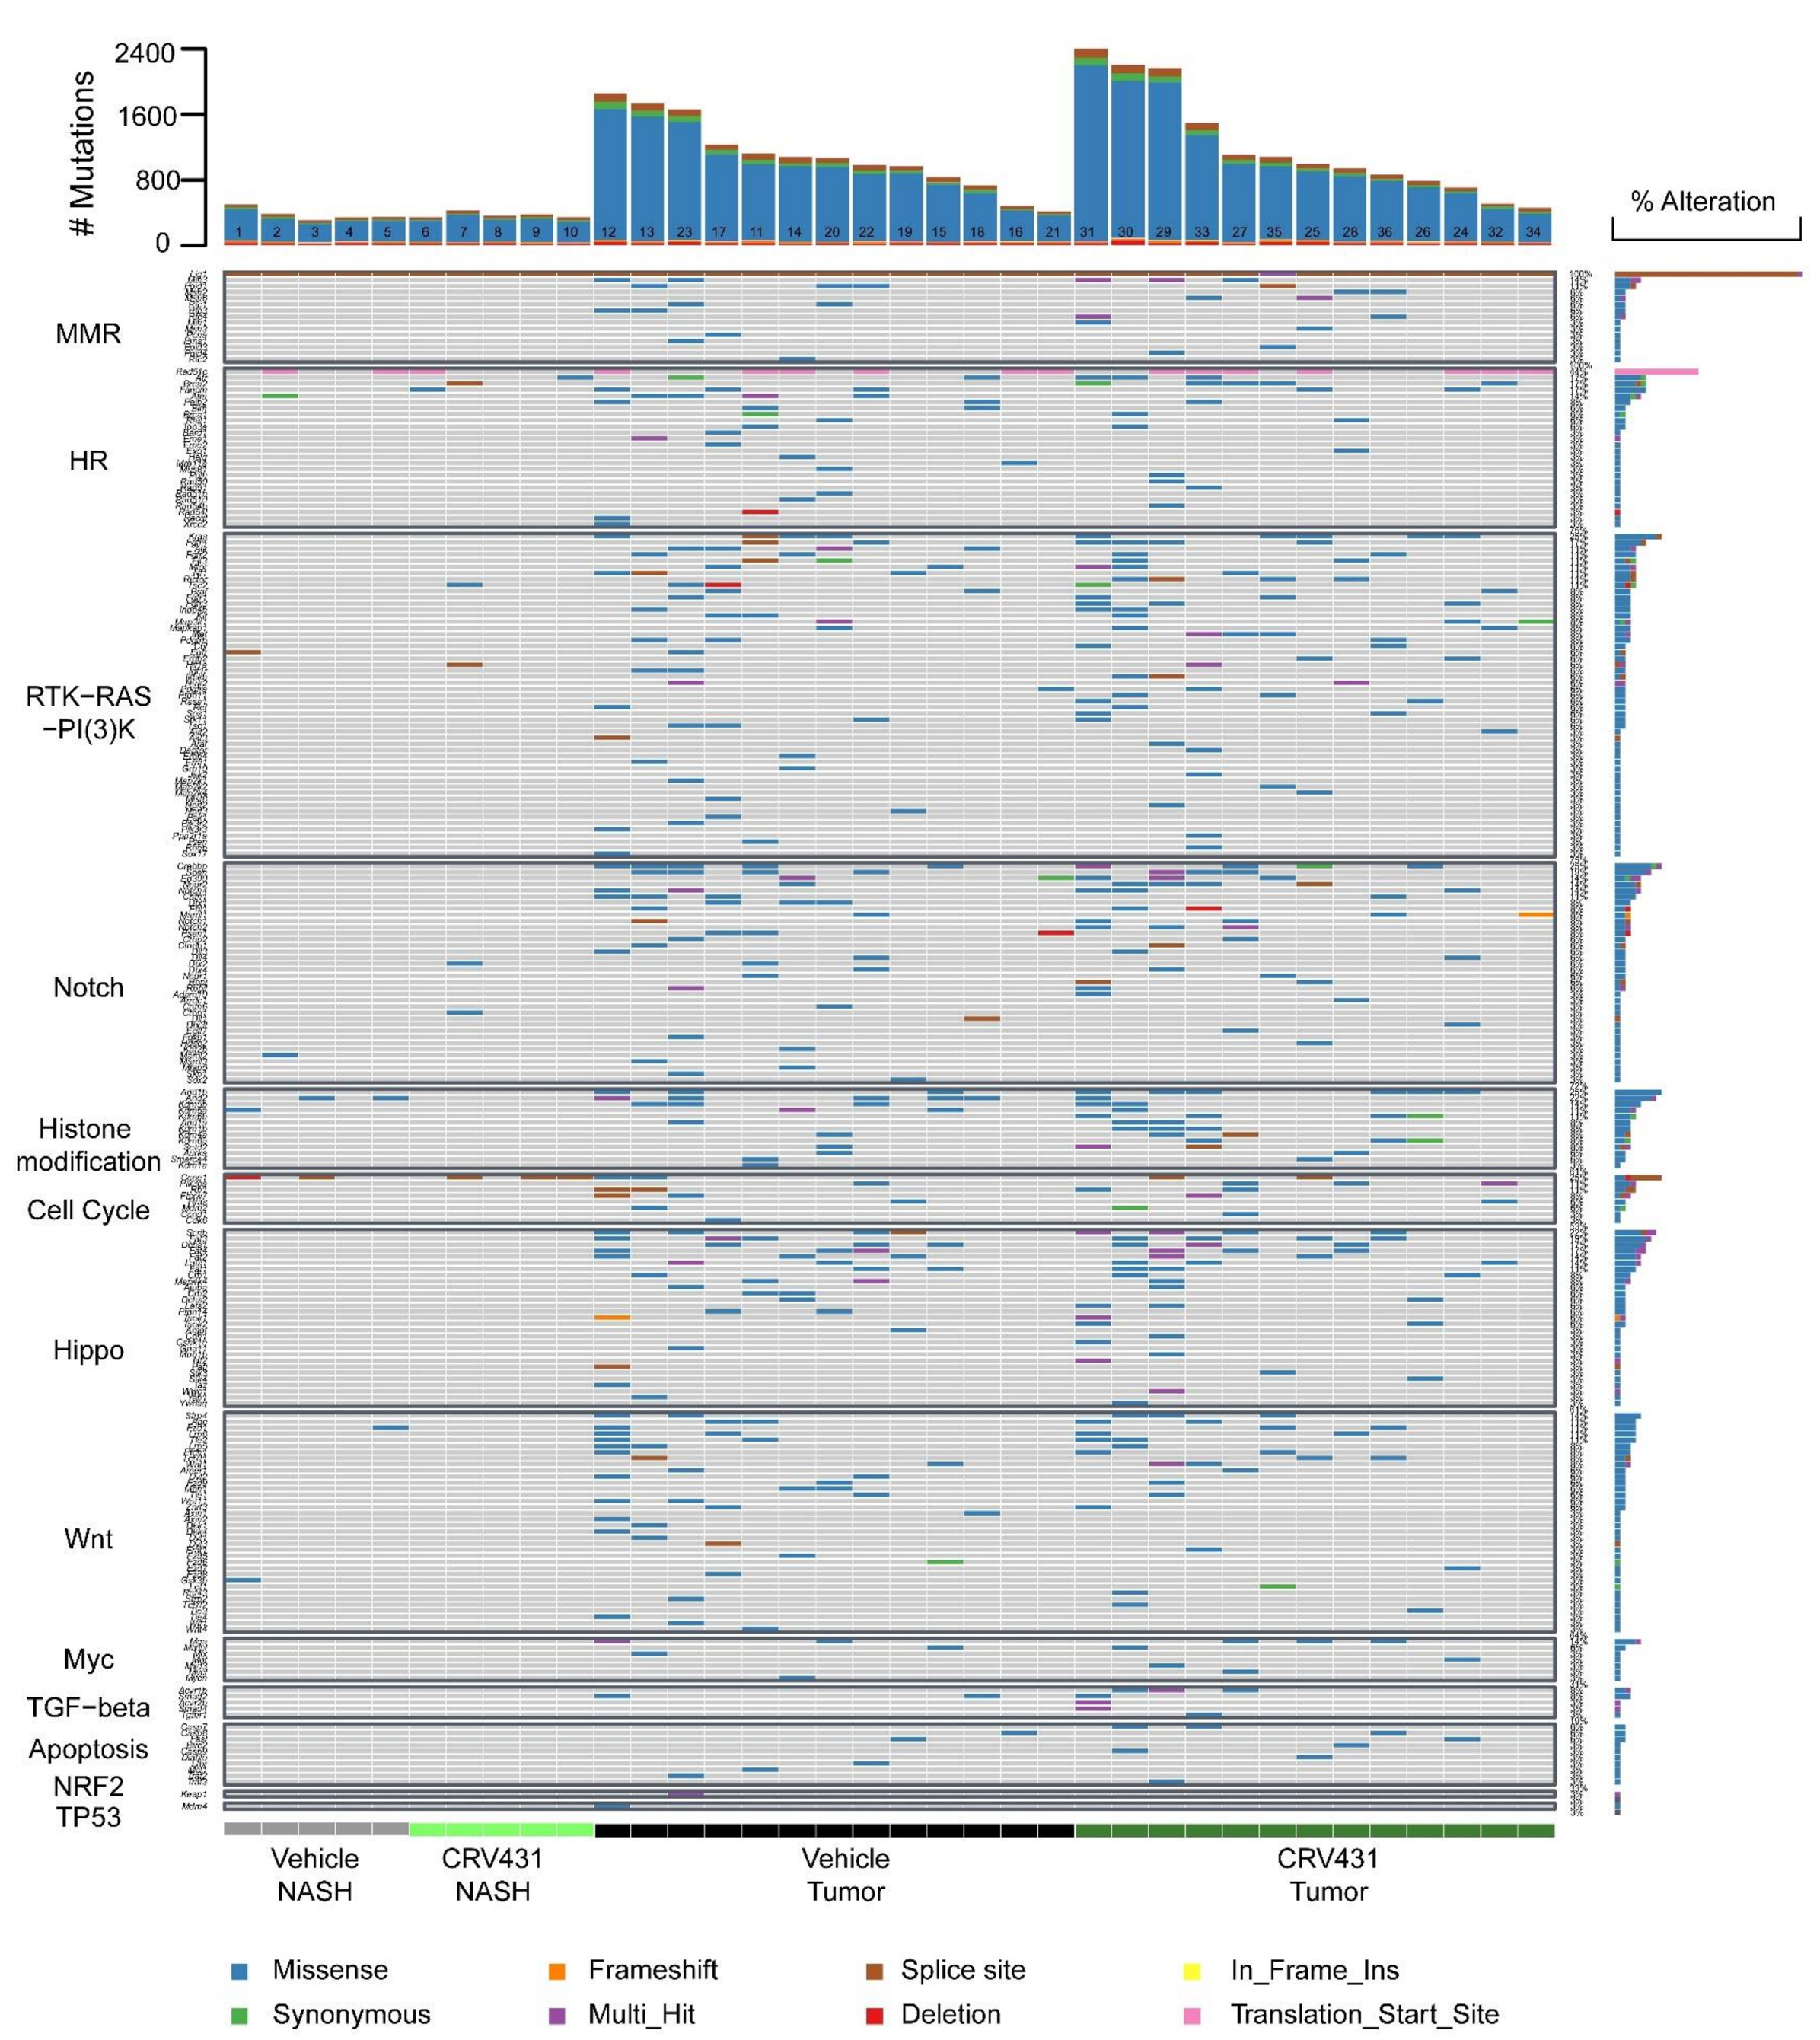
Mutations
% Alteration
MMR
HR
RTK−RAS −PI(3)K
Notch
Histone modification
Cell Cycle
Hippo
Wnt
Myc
TGF−beta
Apoptosis
NRF2
TP53
Vehicle NASH
CRV431 NASH
Vehicle Tumor
CRV431 Tumor
Missense
Synonymous
Frameshift
Multi_Hit
Splice site
Deletion
In_Frame_Ins
Translation_Start_Site

"Normalizing" Effect of Rencoflstat on Metabolomic and Transcriptomic Tumor Signatures

24% of metabolites and 17% of genes were significantly altered in expression in Vehicle tumors compared to Vehicle NASH tissue. Rencofilstat reversed the changes for **47%** of the tumor-signature metabolites and **59%** of the tumor-signature genes (P>0.05 vs Vehicle NASH group).

Relative Abundance of Tumor-Dysregulated Metabolites

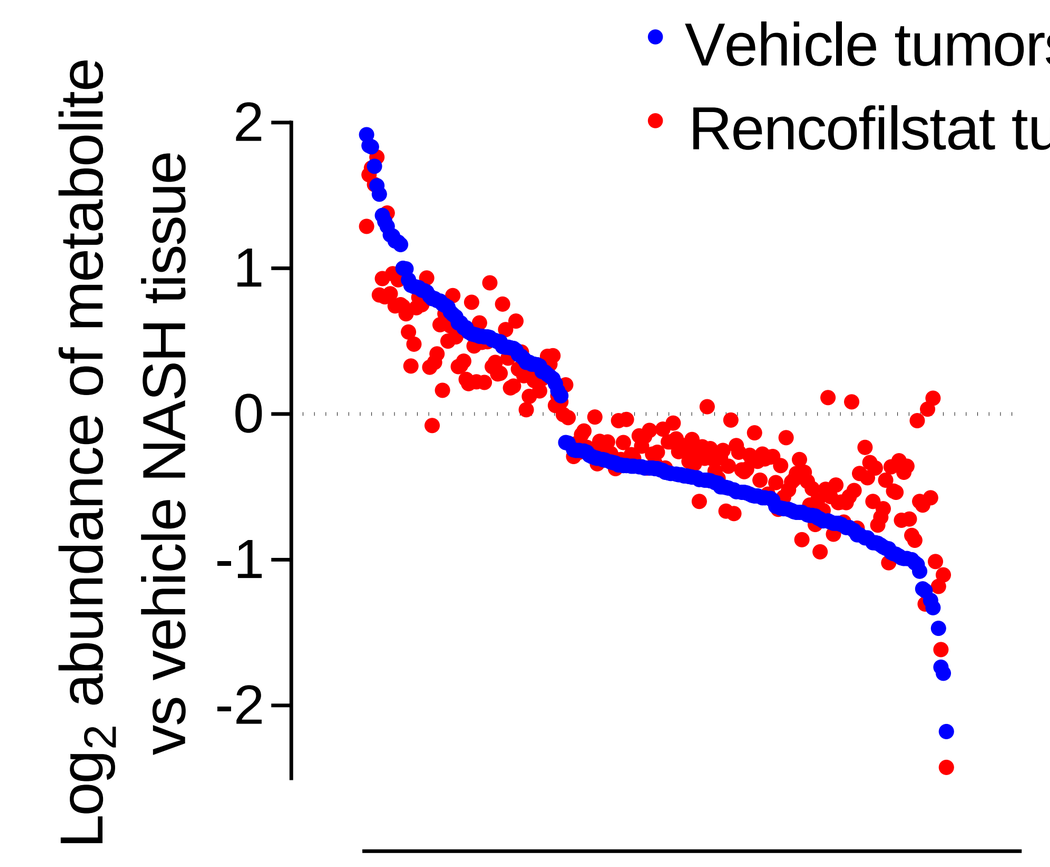
Vehicle tumors
Rencofilstat tumors
Log2 abundance of metabolite vs vehicle NASH tissue

222/907 metabolites (24%) dysregulated in Vehicle tumors

Relative Expression of Tumor-Dysregulated Genes

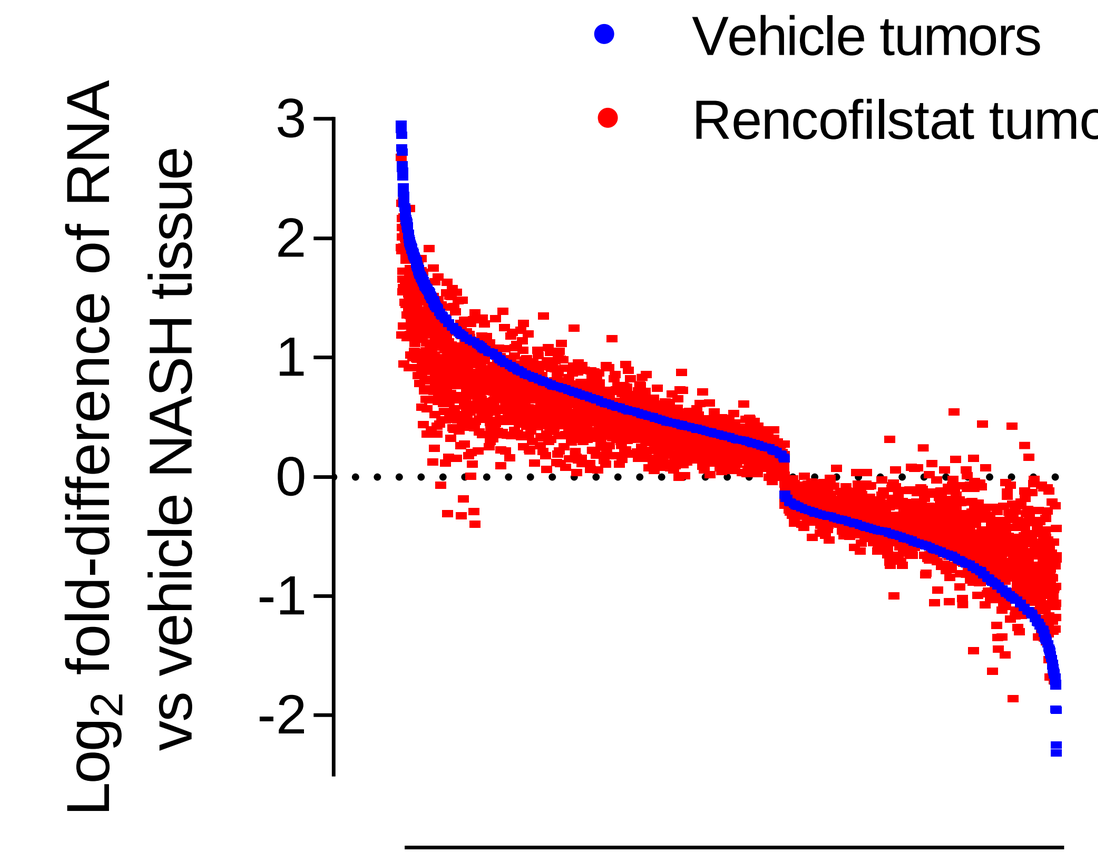
Vehicle tumors
Rencofilstat tumors
Log2 fold-difference of RNA vs vehicle NASH tissue

3,556 / 20,398 genes (17%) dysregulated in Vehicle tumors

Hierarchical Clustering of Metabolites and Gene Expression

Hierarchical clustering identified metabolites and genes significantly altered by rencofilstat in tumor and NASH tissue, including the oncometabolite, **fumarate**, and many fumarate-correlated cancer-driving genes.

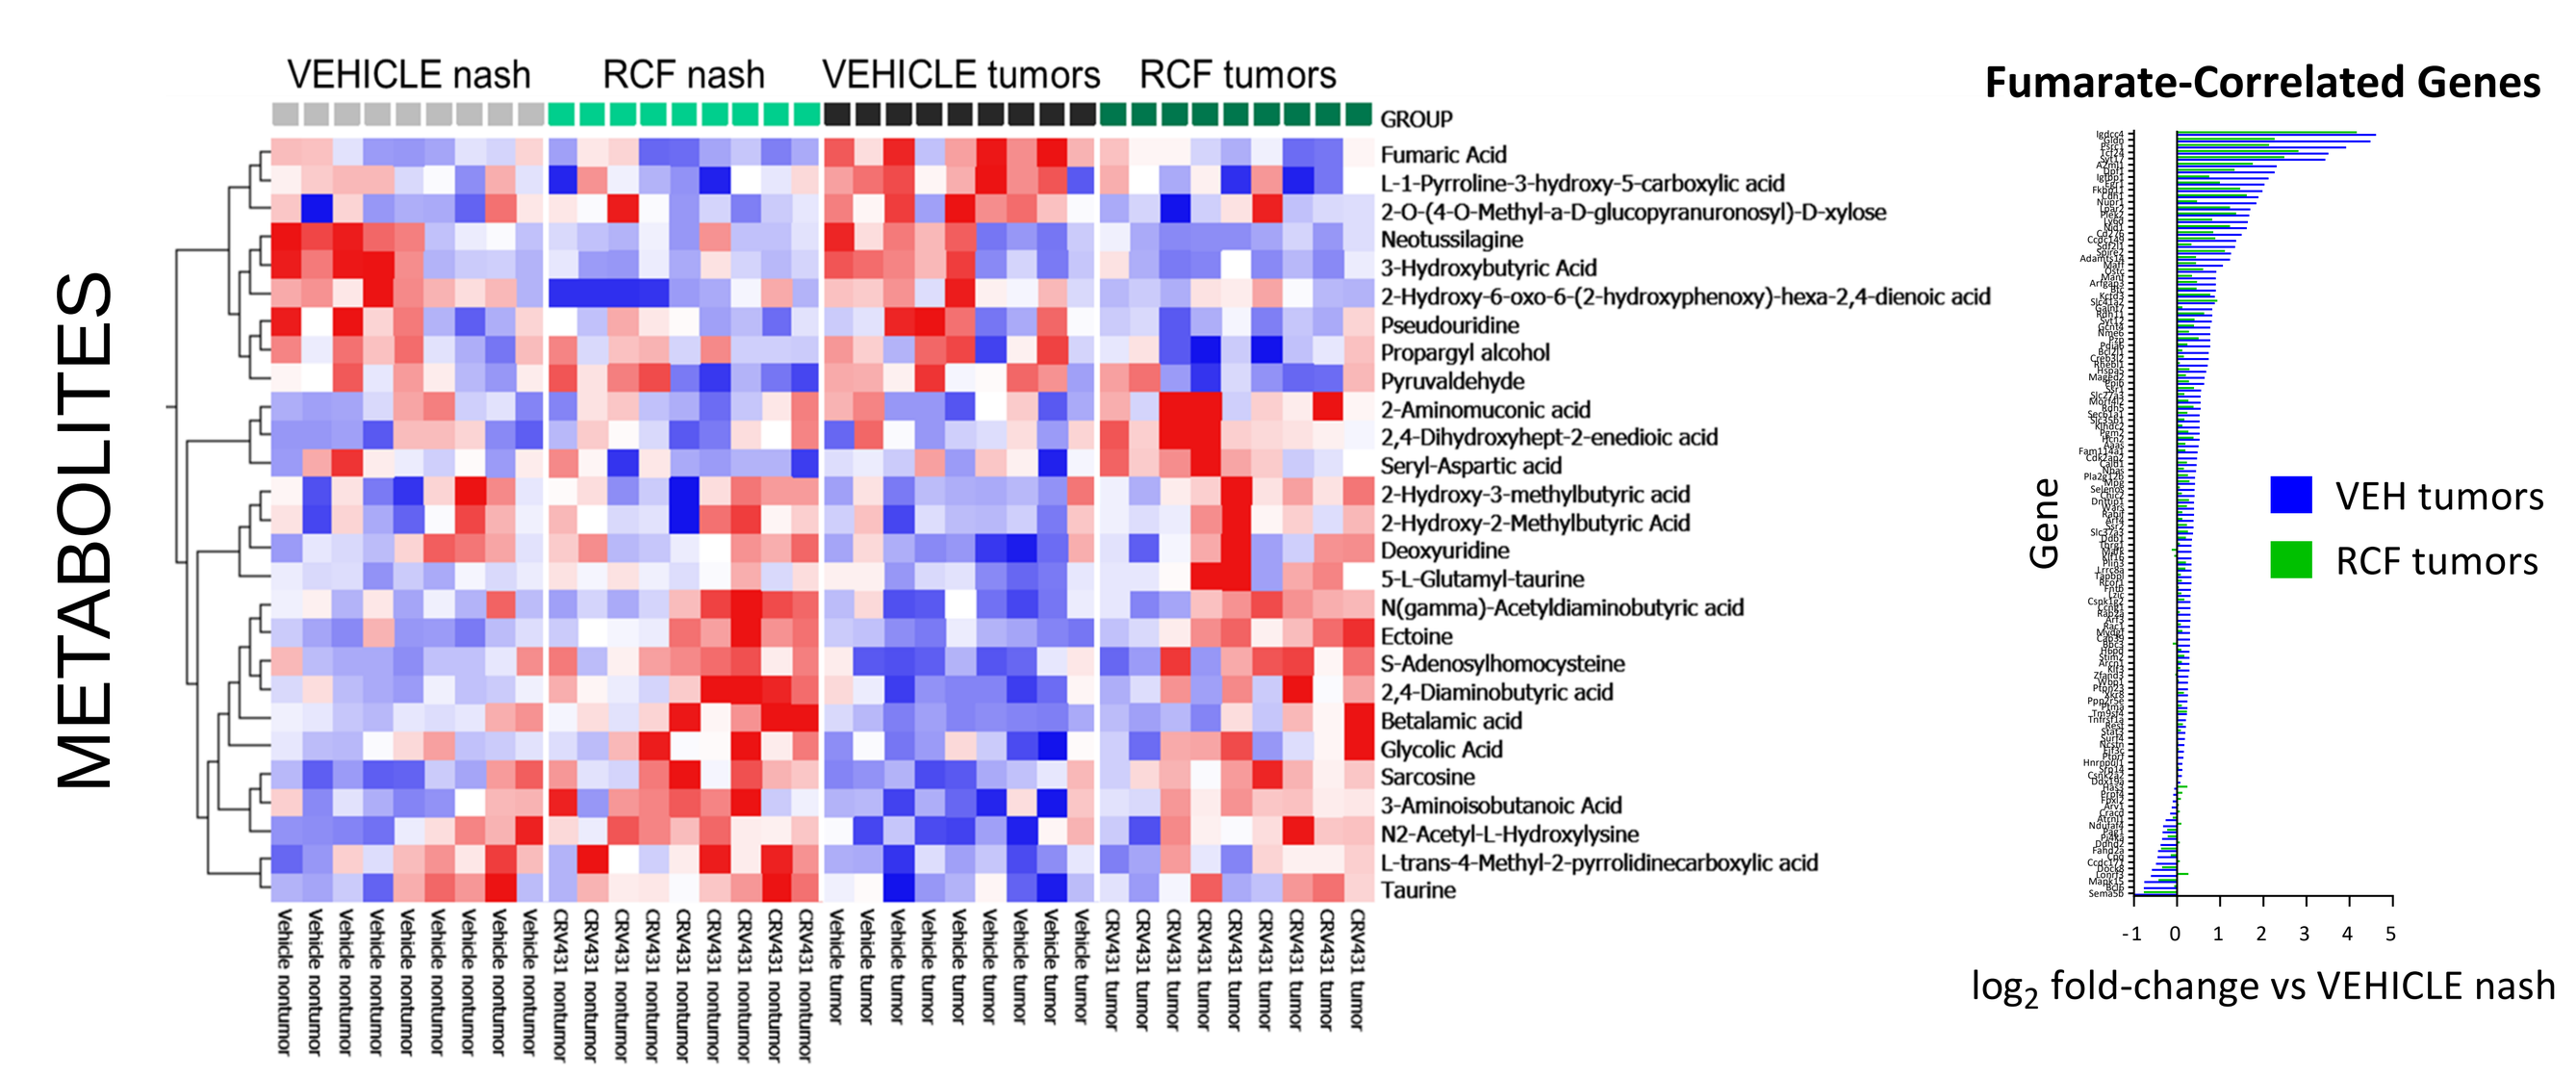
METABOLITES
VEHICLE nash
RCF nash
VEHICLE tumors
RCF tumors
Fumaric Acid
Fumarate-Correlated Genes
VEH tumors
RCF tumors
log2 fold-change vs VEHICLE nash

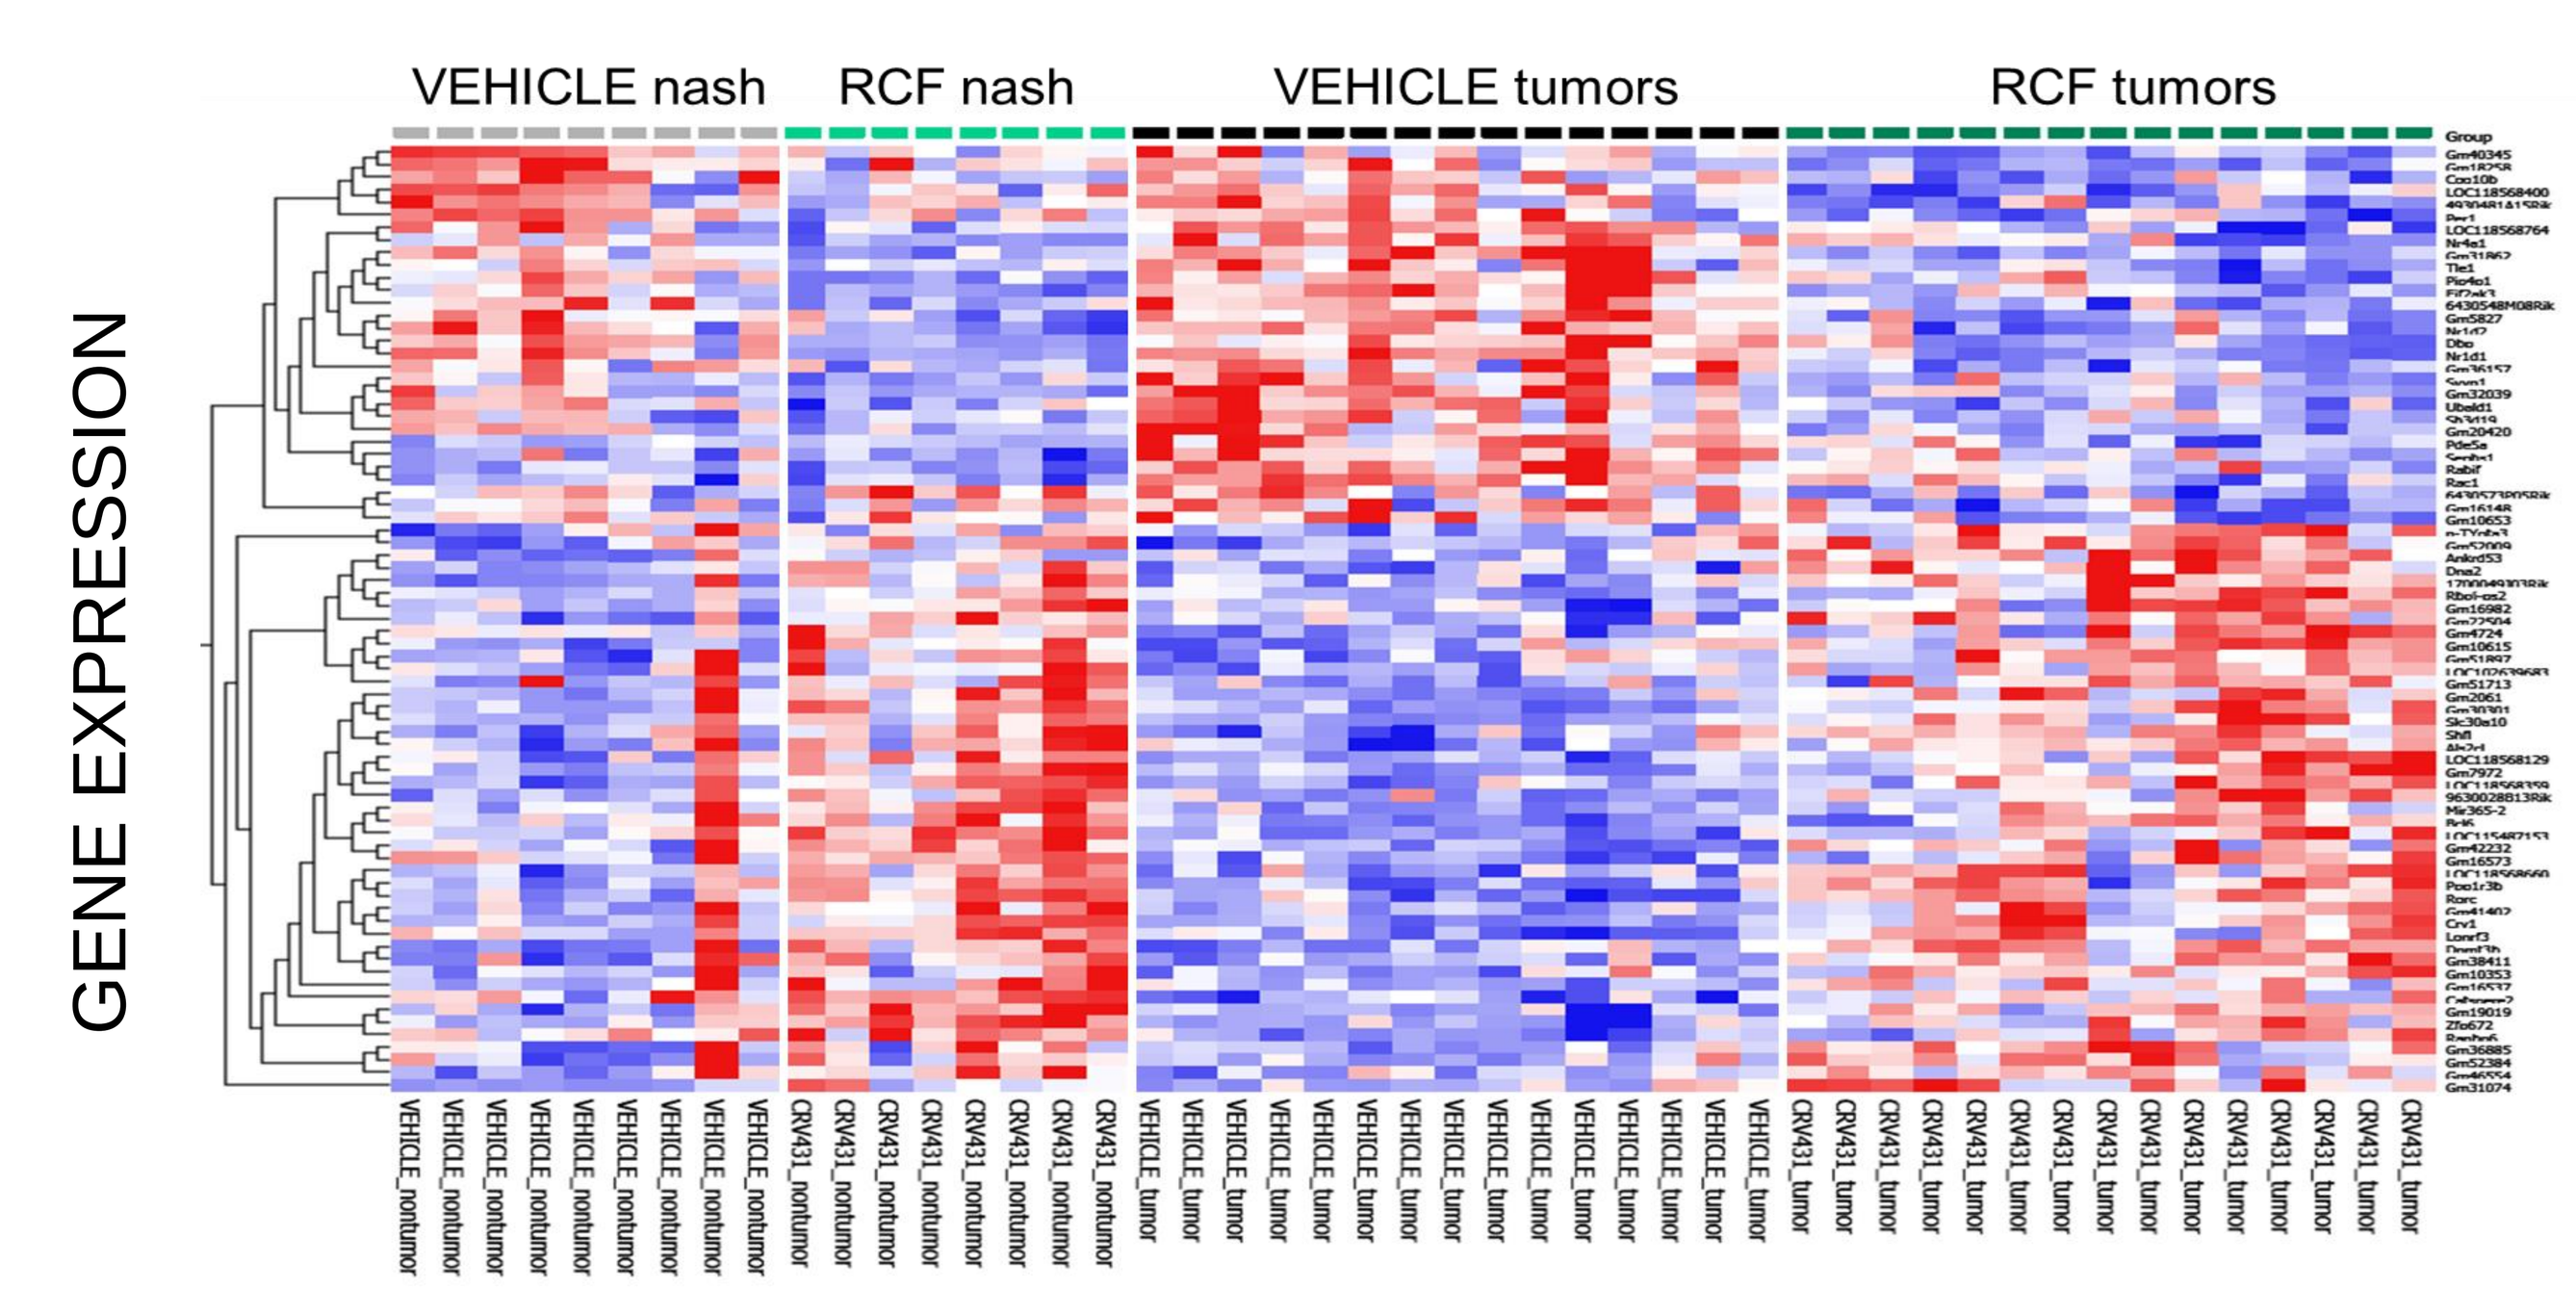
GENE EXPRESSION
VEHICLE nash
RCF nash
VEHICLE tumors
RCF tumors

Transcriptomic Pathway Analysis

Rencofilstat effects on gene expression associated significantly with attenuated oncogenic signaling, as in "**Hepatocellular Carcinoma**".

ADVAITA iPATHWAY GUIDE: "Hepatocellular Carcinoma"

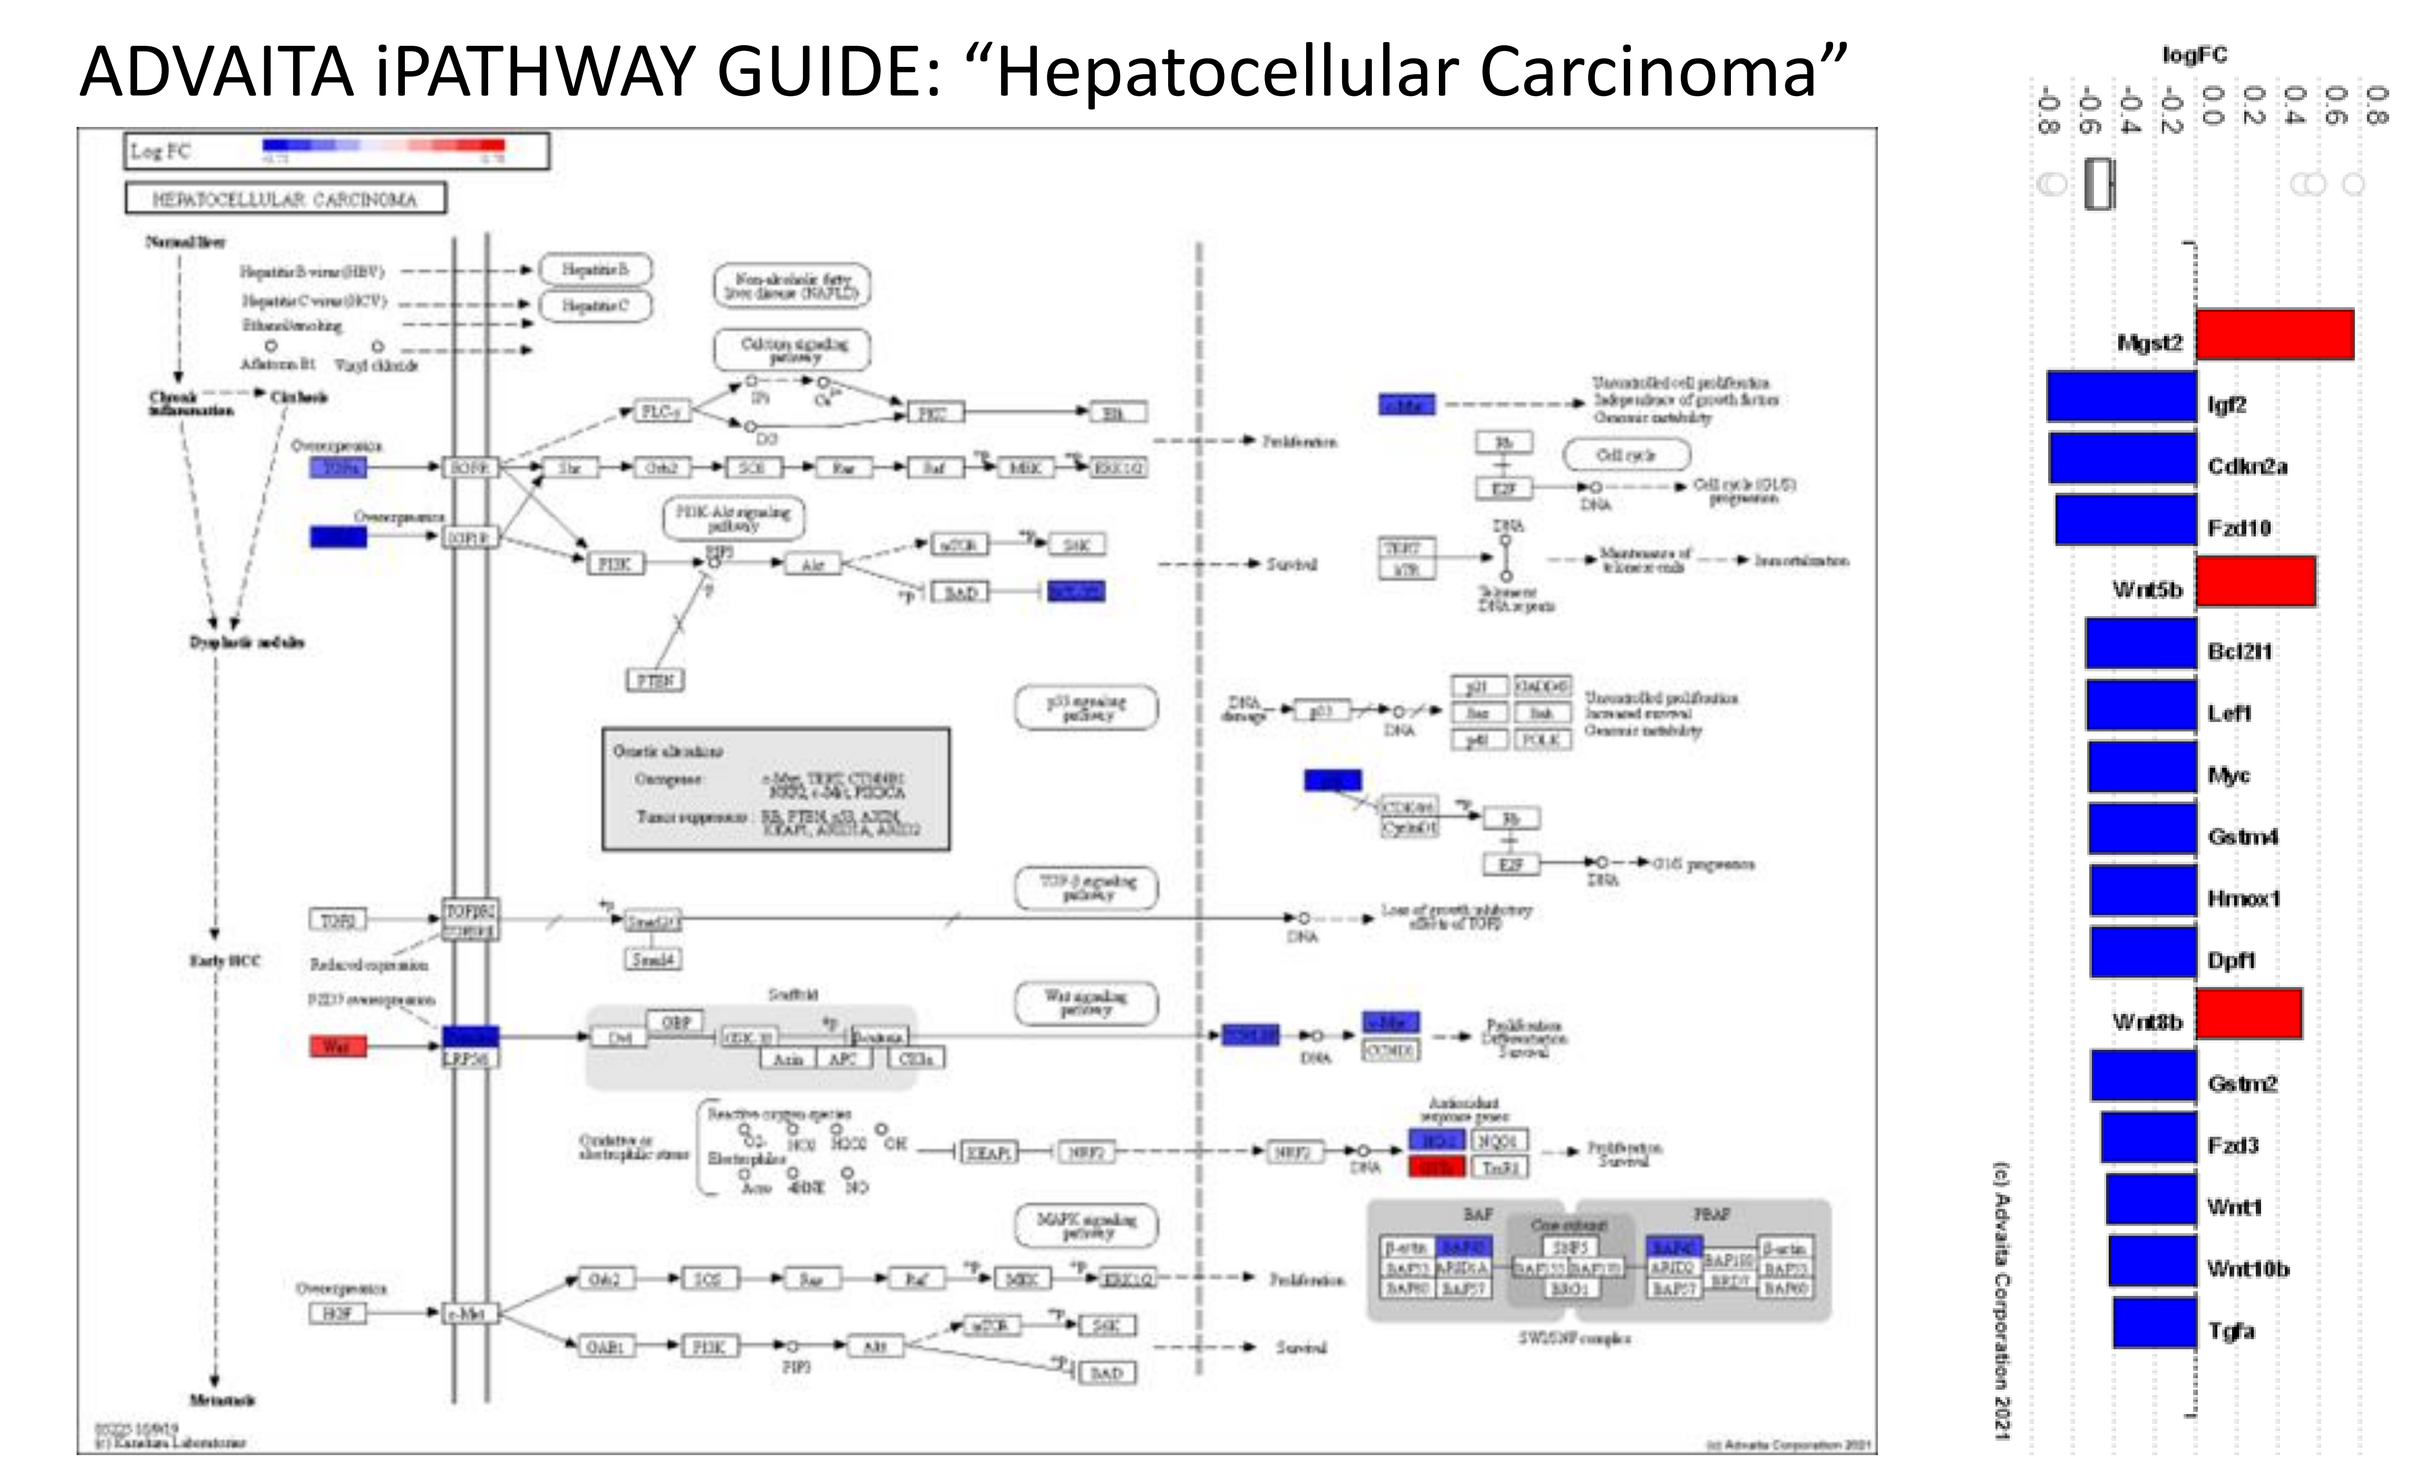
logFC
Mgst2
Igf2
Cdkn2a
Fzd10
Wnt5b
Bcl2l1
Lef1
Myc
Gstm4
Hmox1
Dpf1
Wnt8b
Gstm2
Fzd3
Wnt1
Wnt10b
Tgfa
(c) Advaita Corporation 2021

Key Changes in Gene Expression

Increased expression of cyclophilins B and C in tumors.

Rencofilstat-induced decrease in cancer-driver and stress-response gene expression in tumors and NASH tissue.

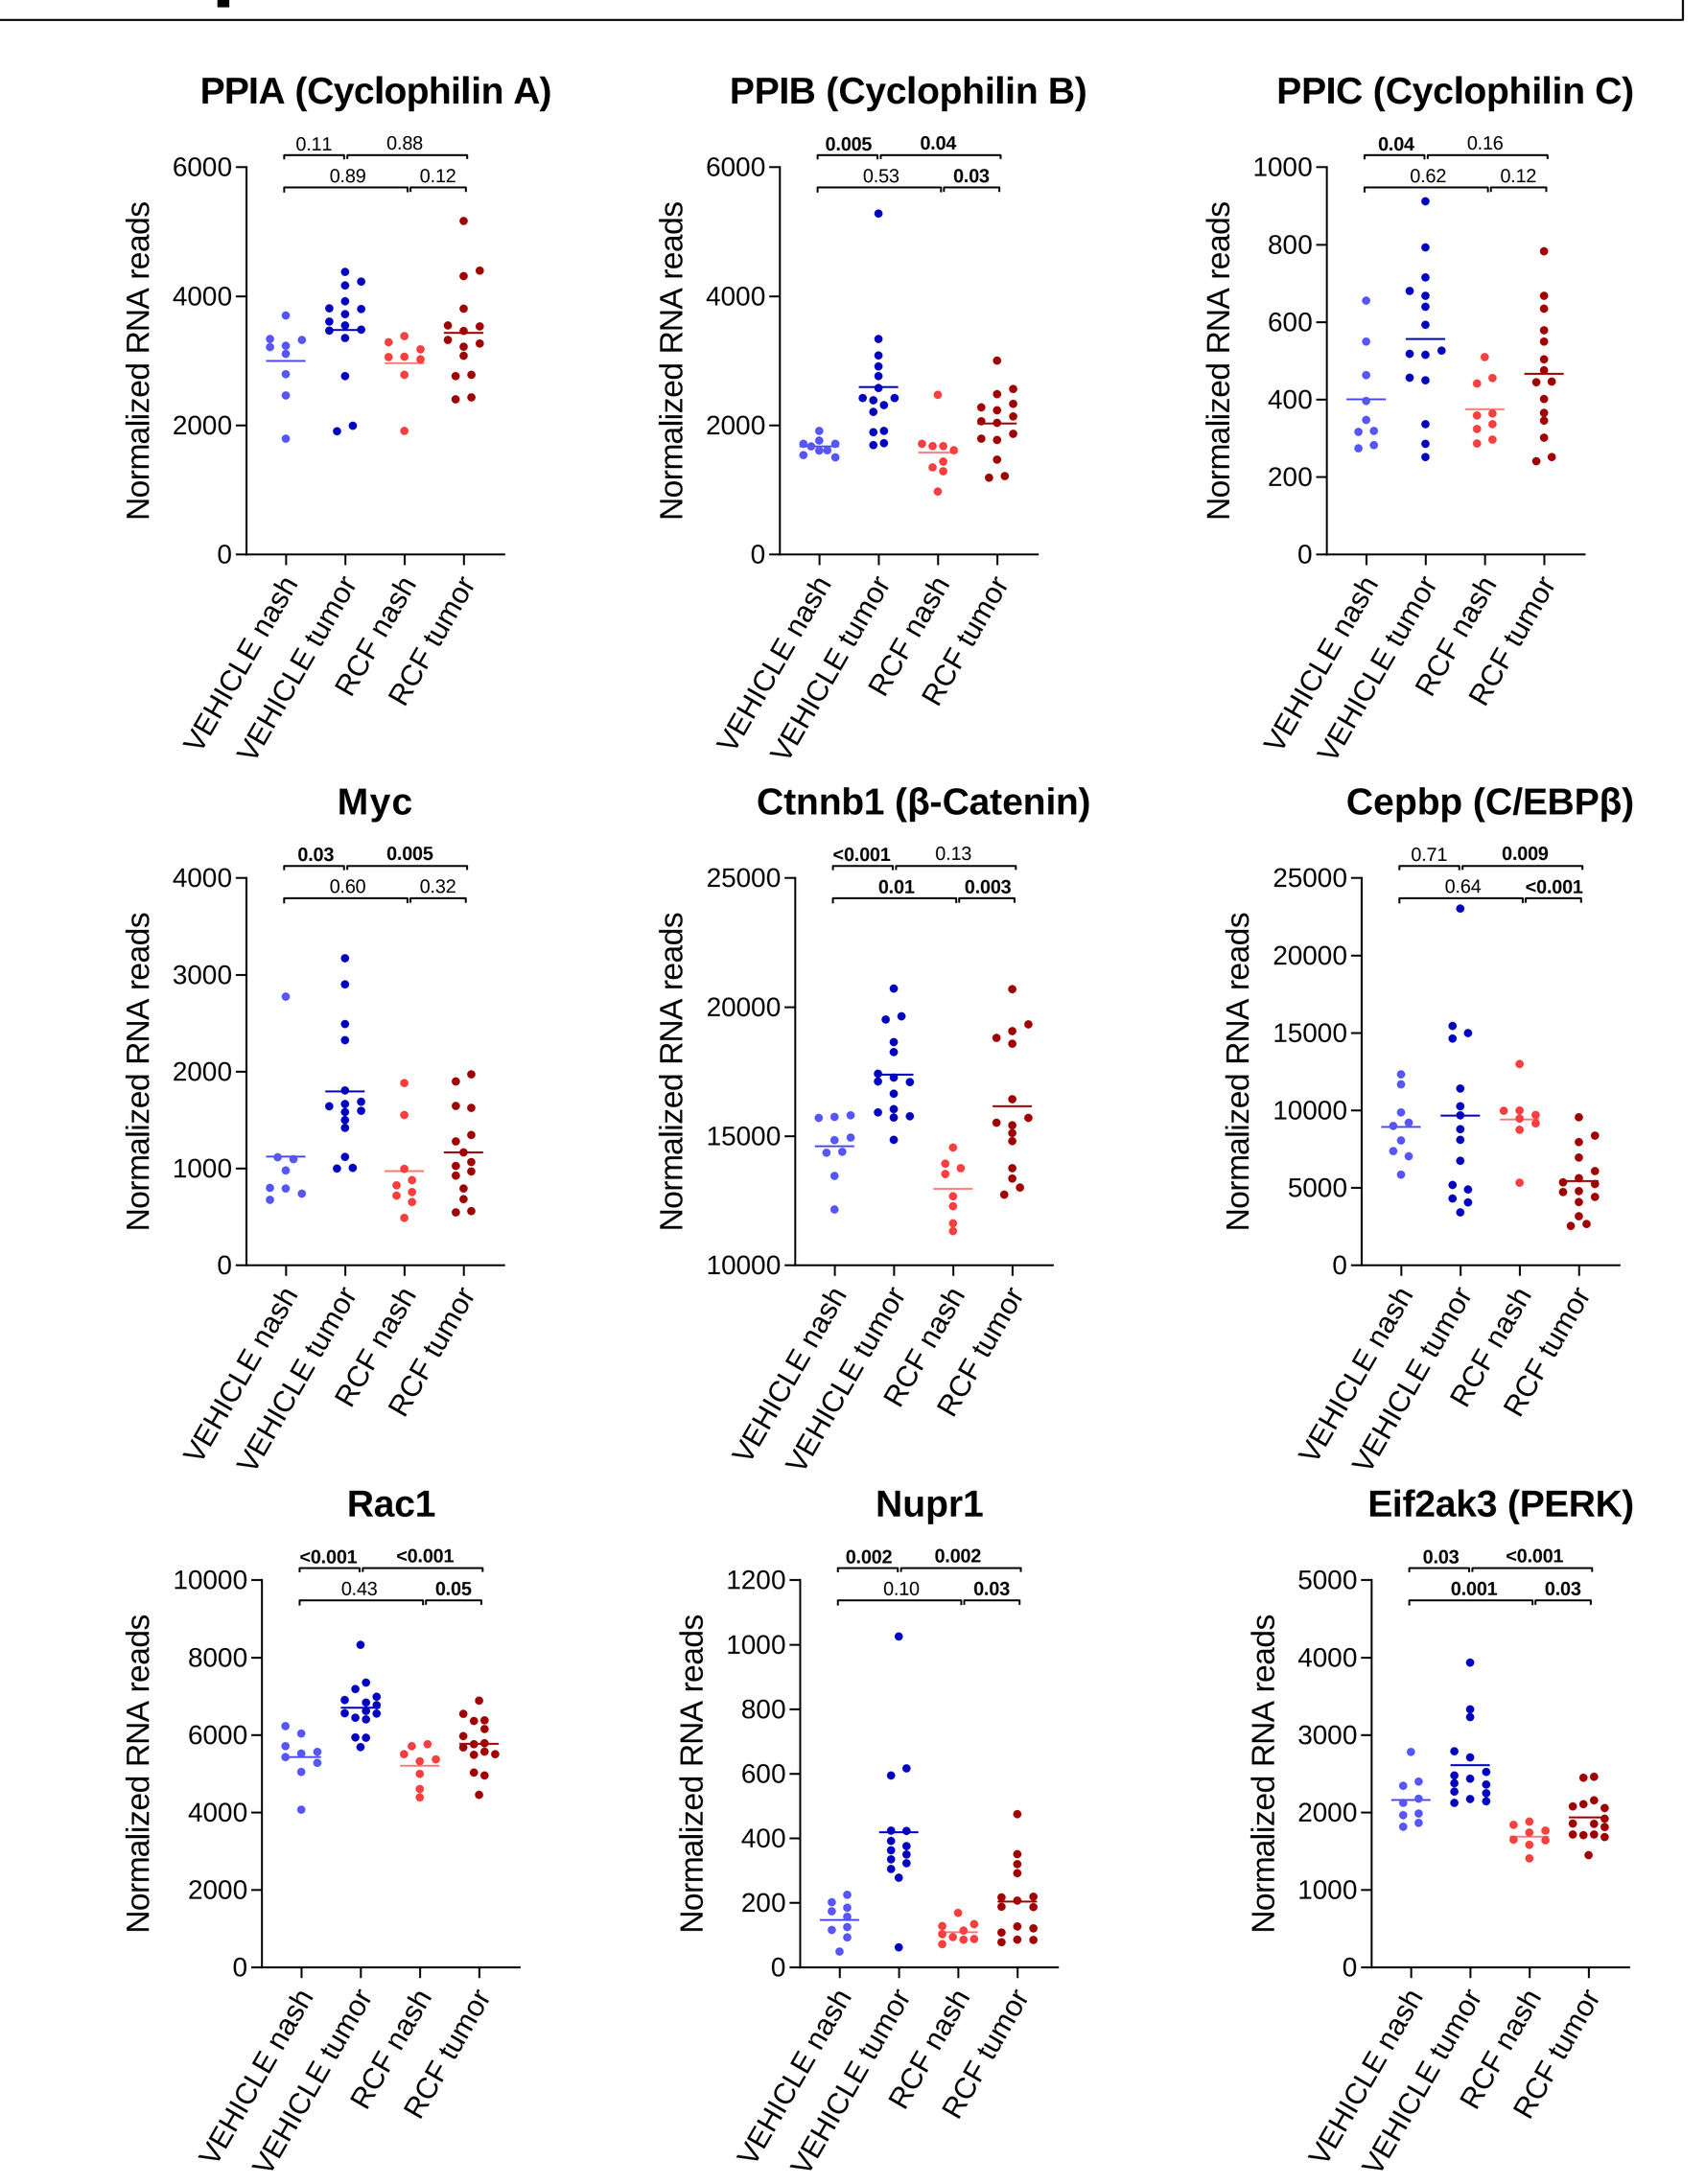
PPIA (Cyclophilin A)
PPIB (Cyclophilin B)
PPIC (Cyclophilin C)
Myc
Ctnnb1 (β-Catenin)
Cepbp (C/EBPβ)
Rac1
Nupr1
Eif2ak3 (PERK)
Normalized RNA reads
VEHICLE nash
VEHICLE tumor
RCF nash
RCF tumor

CONCLUSIONS

Rencofilstat decreased liver tumor burden in a chemical/Western diet model of HCC.

The tumor-reducing effect of rencofilstat was associated with attenuation of oncogenic pathways found in human HCC, and with significant normalization of the tumor gene expression signature.