CRV431 decreases diet- and chemical-driven fibrosis in livers of mice

Joseph Kuo[1], **Erika Cabrera**[1], **Patrick Mayo**[2], **Daren Ure**[2], **Robert Foster**[2], **and Philippe Gallay**[1]

[1]Department of Immunology & Microbiology, The Scripps Research Institute, La Jolla, California 92037, USA; [2]Hepion Pharmaceuticals, Edison, New Jersey, 08837, USA.

BACKGROUND

Liver fibrosis

- Prevalent pathological feature of chronic hepatic injuries
- Accumulation of collagen with inflammation and scarring as clinical sign
- Limited treatment options available for human patients
- Cyclophilins, a family of proteins, has been shown to contribute to the development of fibrosis and later stages (hepatocellular carcinoma)

CRV431

- Pan-cyclophilin inhibitor showing reduction of collagen deposition in CRV431-treated mice with damaged pancreatic cells and nourished with high-fat diet
- Same trend observed in CRV431-treated mice exposed to carbon tetrachloride (CCL_4)

Aim: Examine if CRV431 affects the development of fibrosis induced by both chemical and diet in male C57BL/6J mice; obeticholic acid (OCA) as comparator

EXPERIMENTAL DESIGN

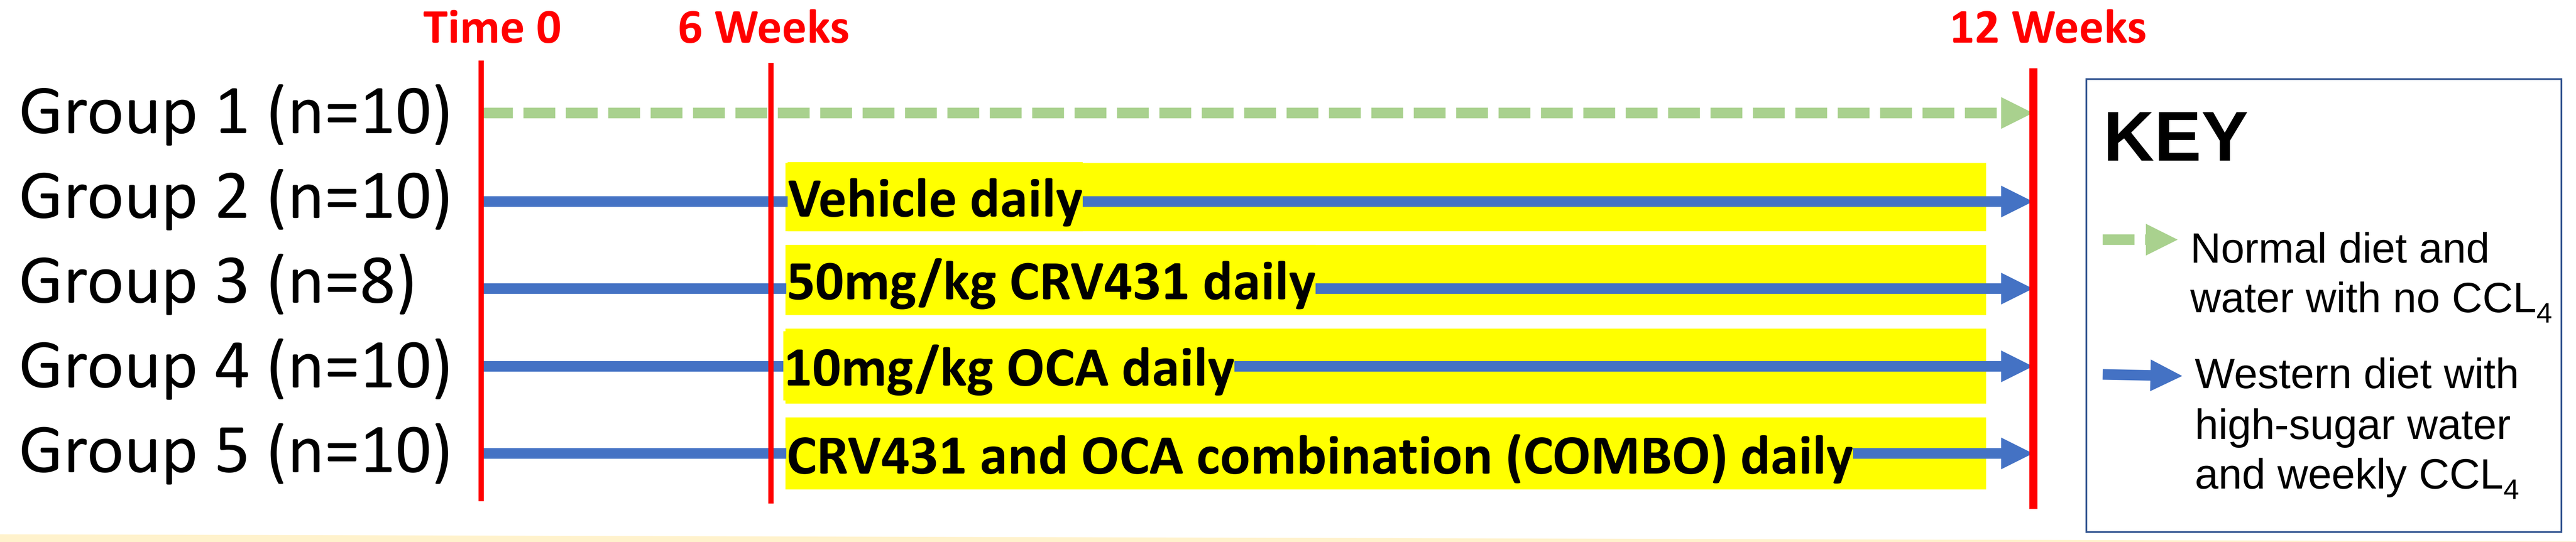
Time 0
6 Weeks
12 Weeks
Group 1 (n=10)
Group 2 (n=10)
Group 3 (n=8)
Group 4 (n=10)
Group 5 (n=10)
Vehicle daily
50mg/kg CRV431 daily
10mg/kg OCA daily
CRV431 and OCA combination (COMBO) daily
KEY
Normal diet and water with no CCL4
Western diet with high-sugar water and weekly CCL4

RESULTS

Group / Detections	Serum					Liver Histology Score		
	Glucose (mg/dL)	Alanine Transaminase (nmole/min/mL)	Aspartate Transaminase (nmole/min/mL)	Alpha Fetoprotein (ng/mL)	Cholesterol (mg/dL)	Inflammation	Steatosis	Ballooning
Negative Control	264.5	9.739	51.35	69.09	202.4	0	0	0
Vehicle	344.4	43.55	79.52	116.2	255.1	3.4	3	2
CRV431	284.1	44.53	79.16	114.7	237.7	3	3	2
OCA	552.5	18.12	64.26	95.45	254.6	2.9	3	2
COMBO	284.6	28.7	76.65	114.6	205.9	2.6	2.8	1.9

CRV431 does not affect glucose and protein levels in serum

- Treatment did not affect levels detected in serum. Bonferroni's multiple comparison tests (two-tailed) were conducted.

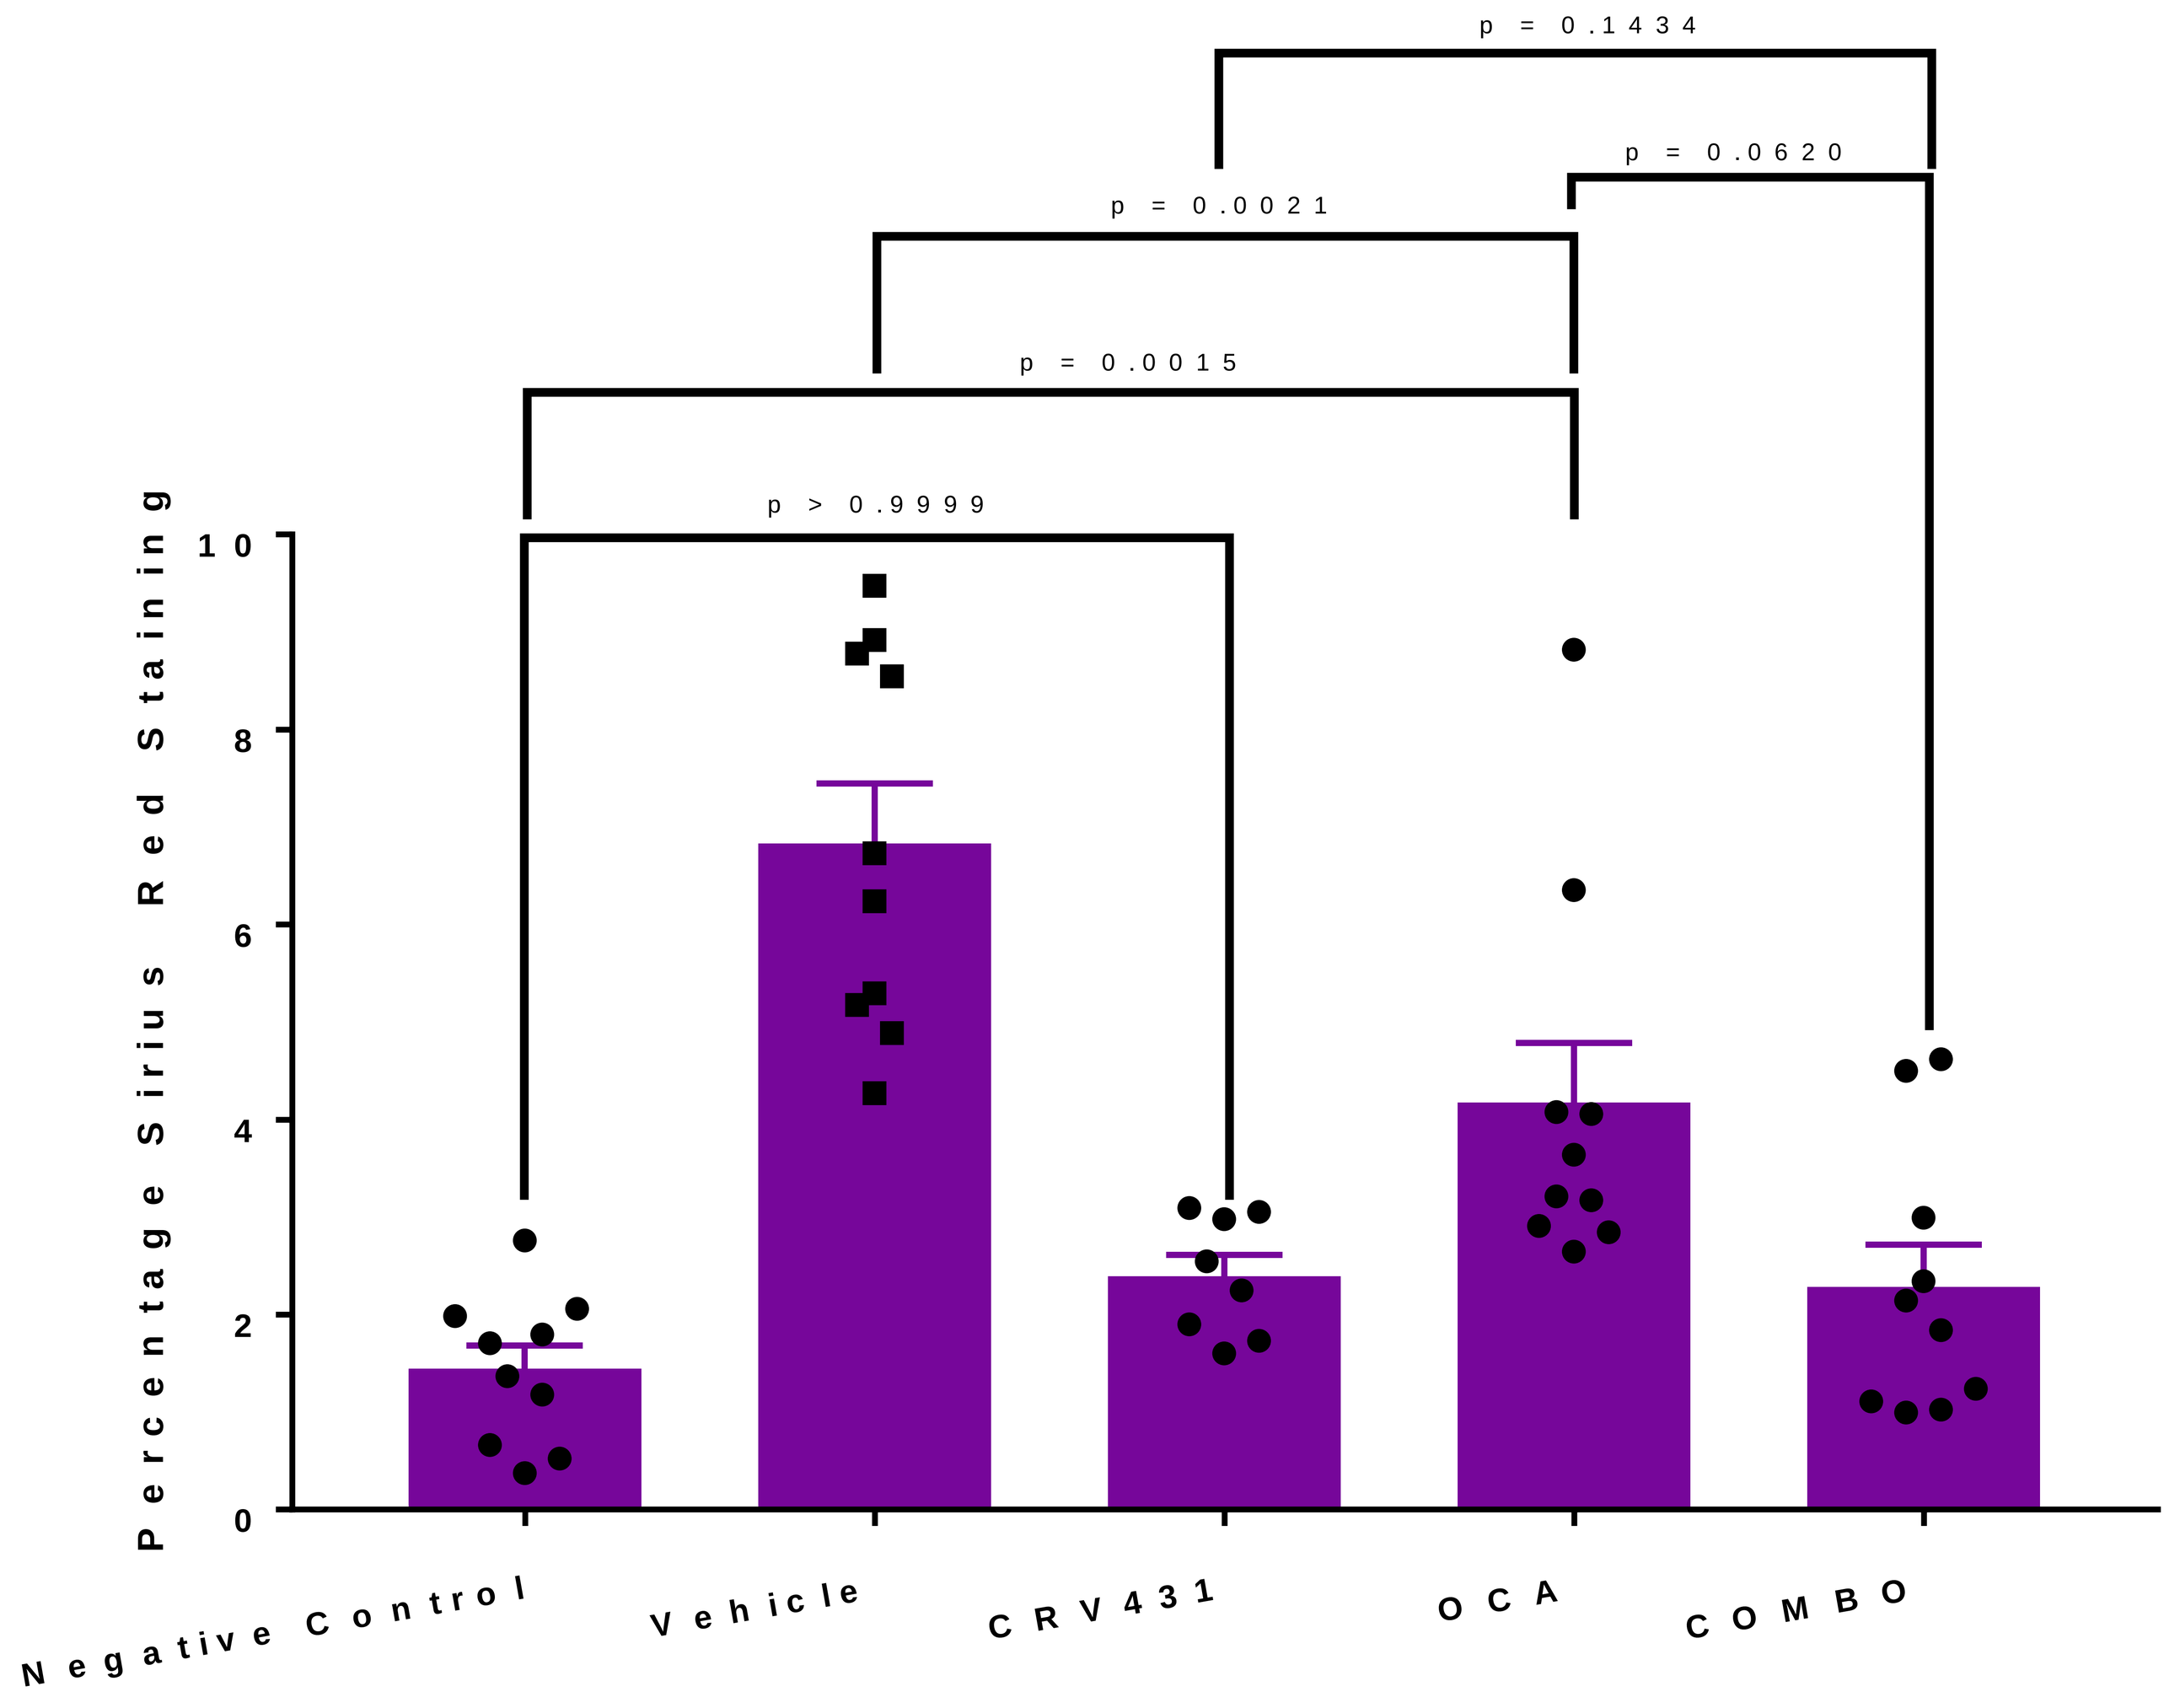
Percentage Sirius Red Staining
0
2
4
6
8
10
p = 0.1434
p = 0.0620
p = 0.0021
p = 0.0015
p > 0.9999
Negative Control
Vehicle
CRV431
OCA
COMBO

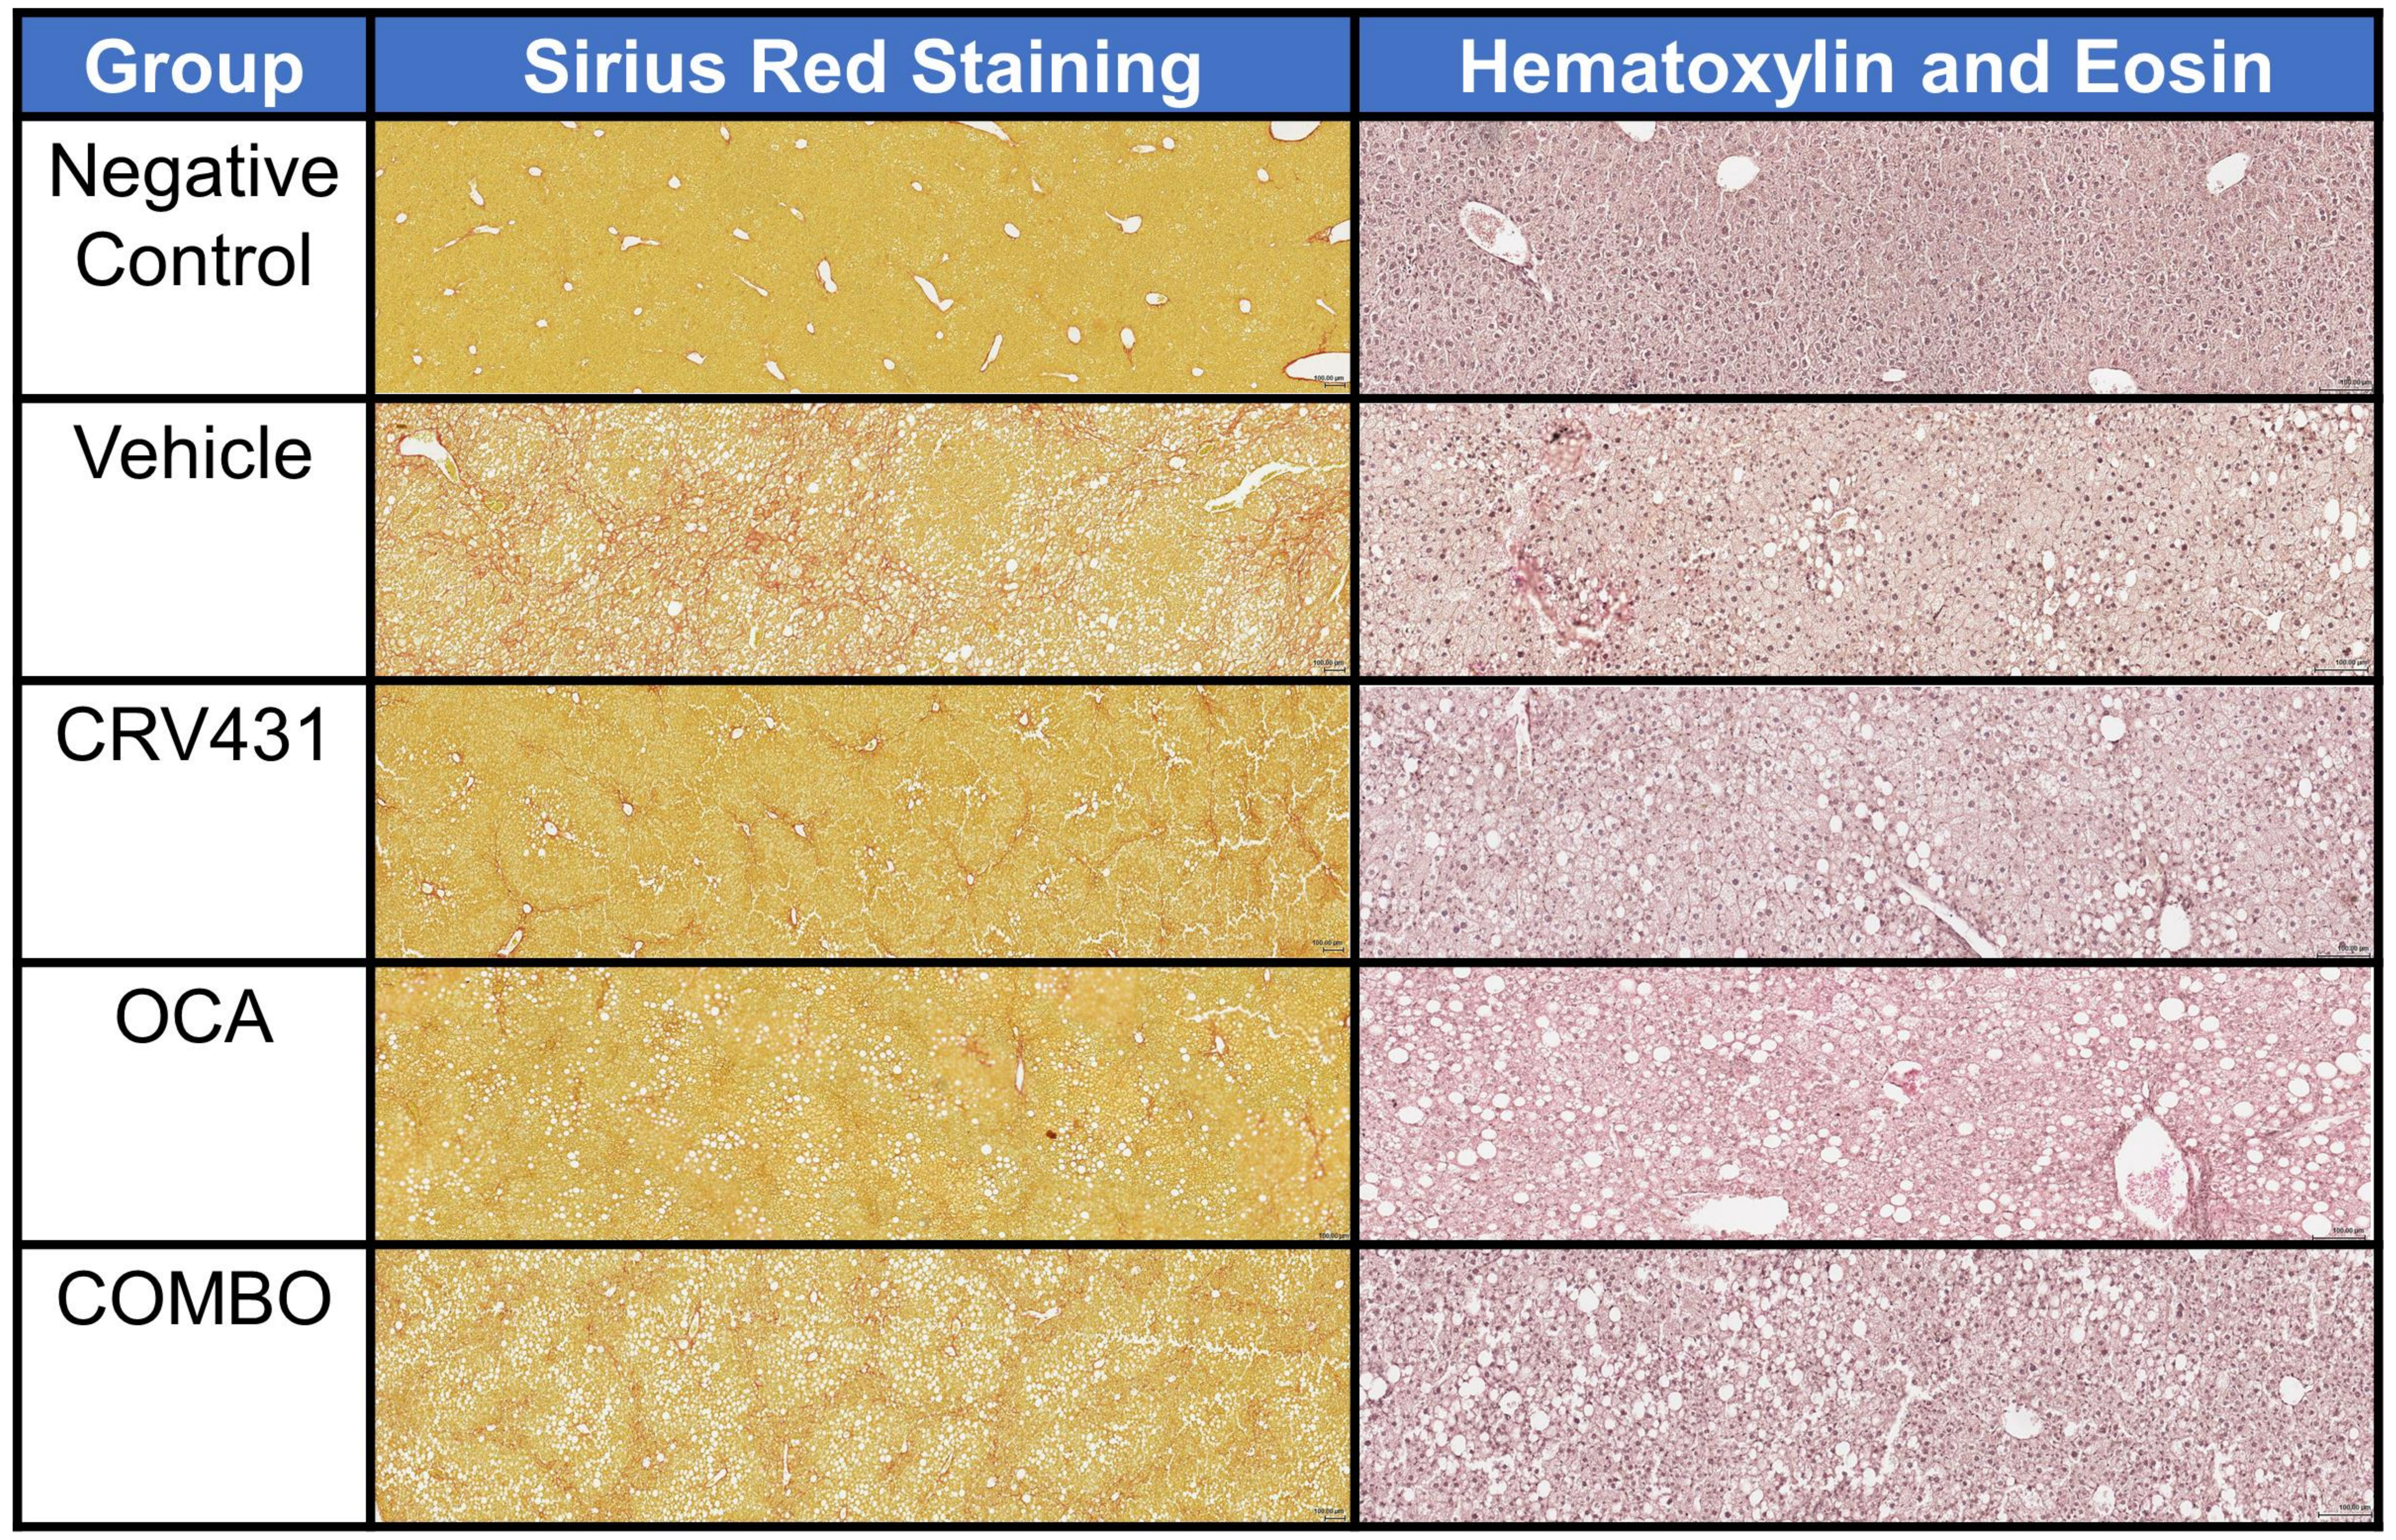
Group
Sirius Red Staining
Hematoxylin and Eosin
Negative Control
Vehicle
CRV431
OCA
COMBO

CRV431 decreases hepatic collagen deposition stained with Sirius Red

- 65% reduction in CRV431-treated mice compared to vehicle controls
- Reduction also seen in OCA, but not to the extent of CRV431

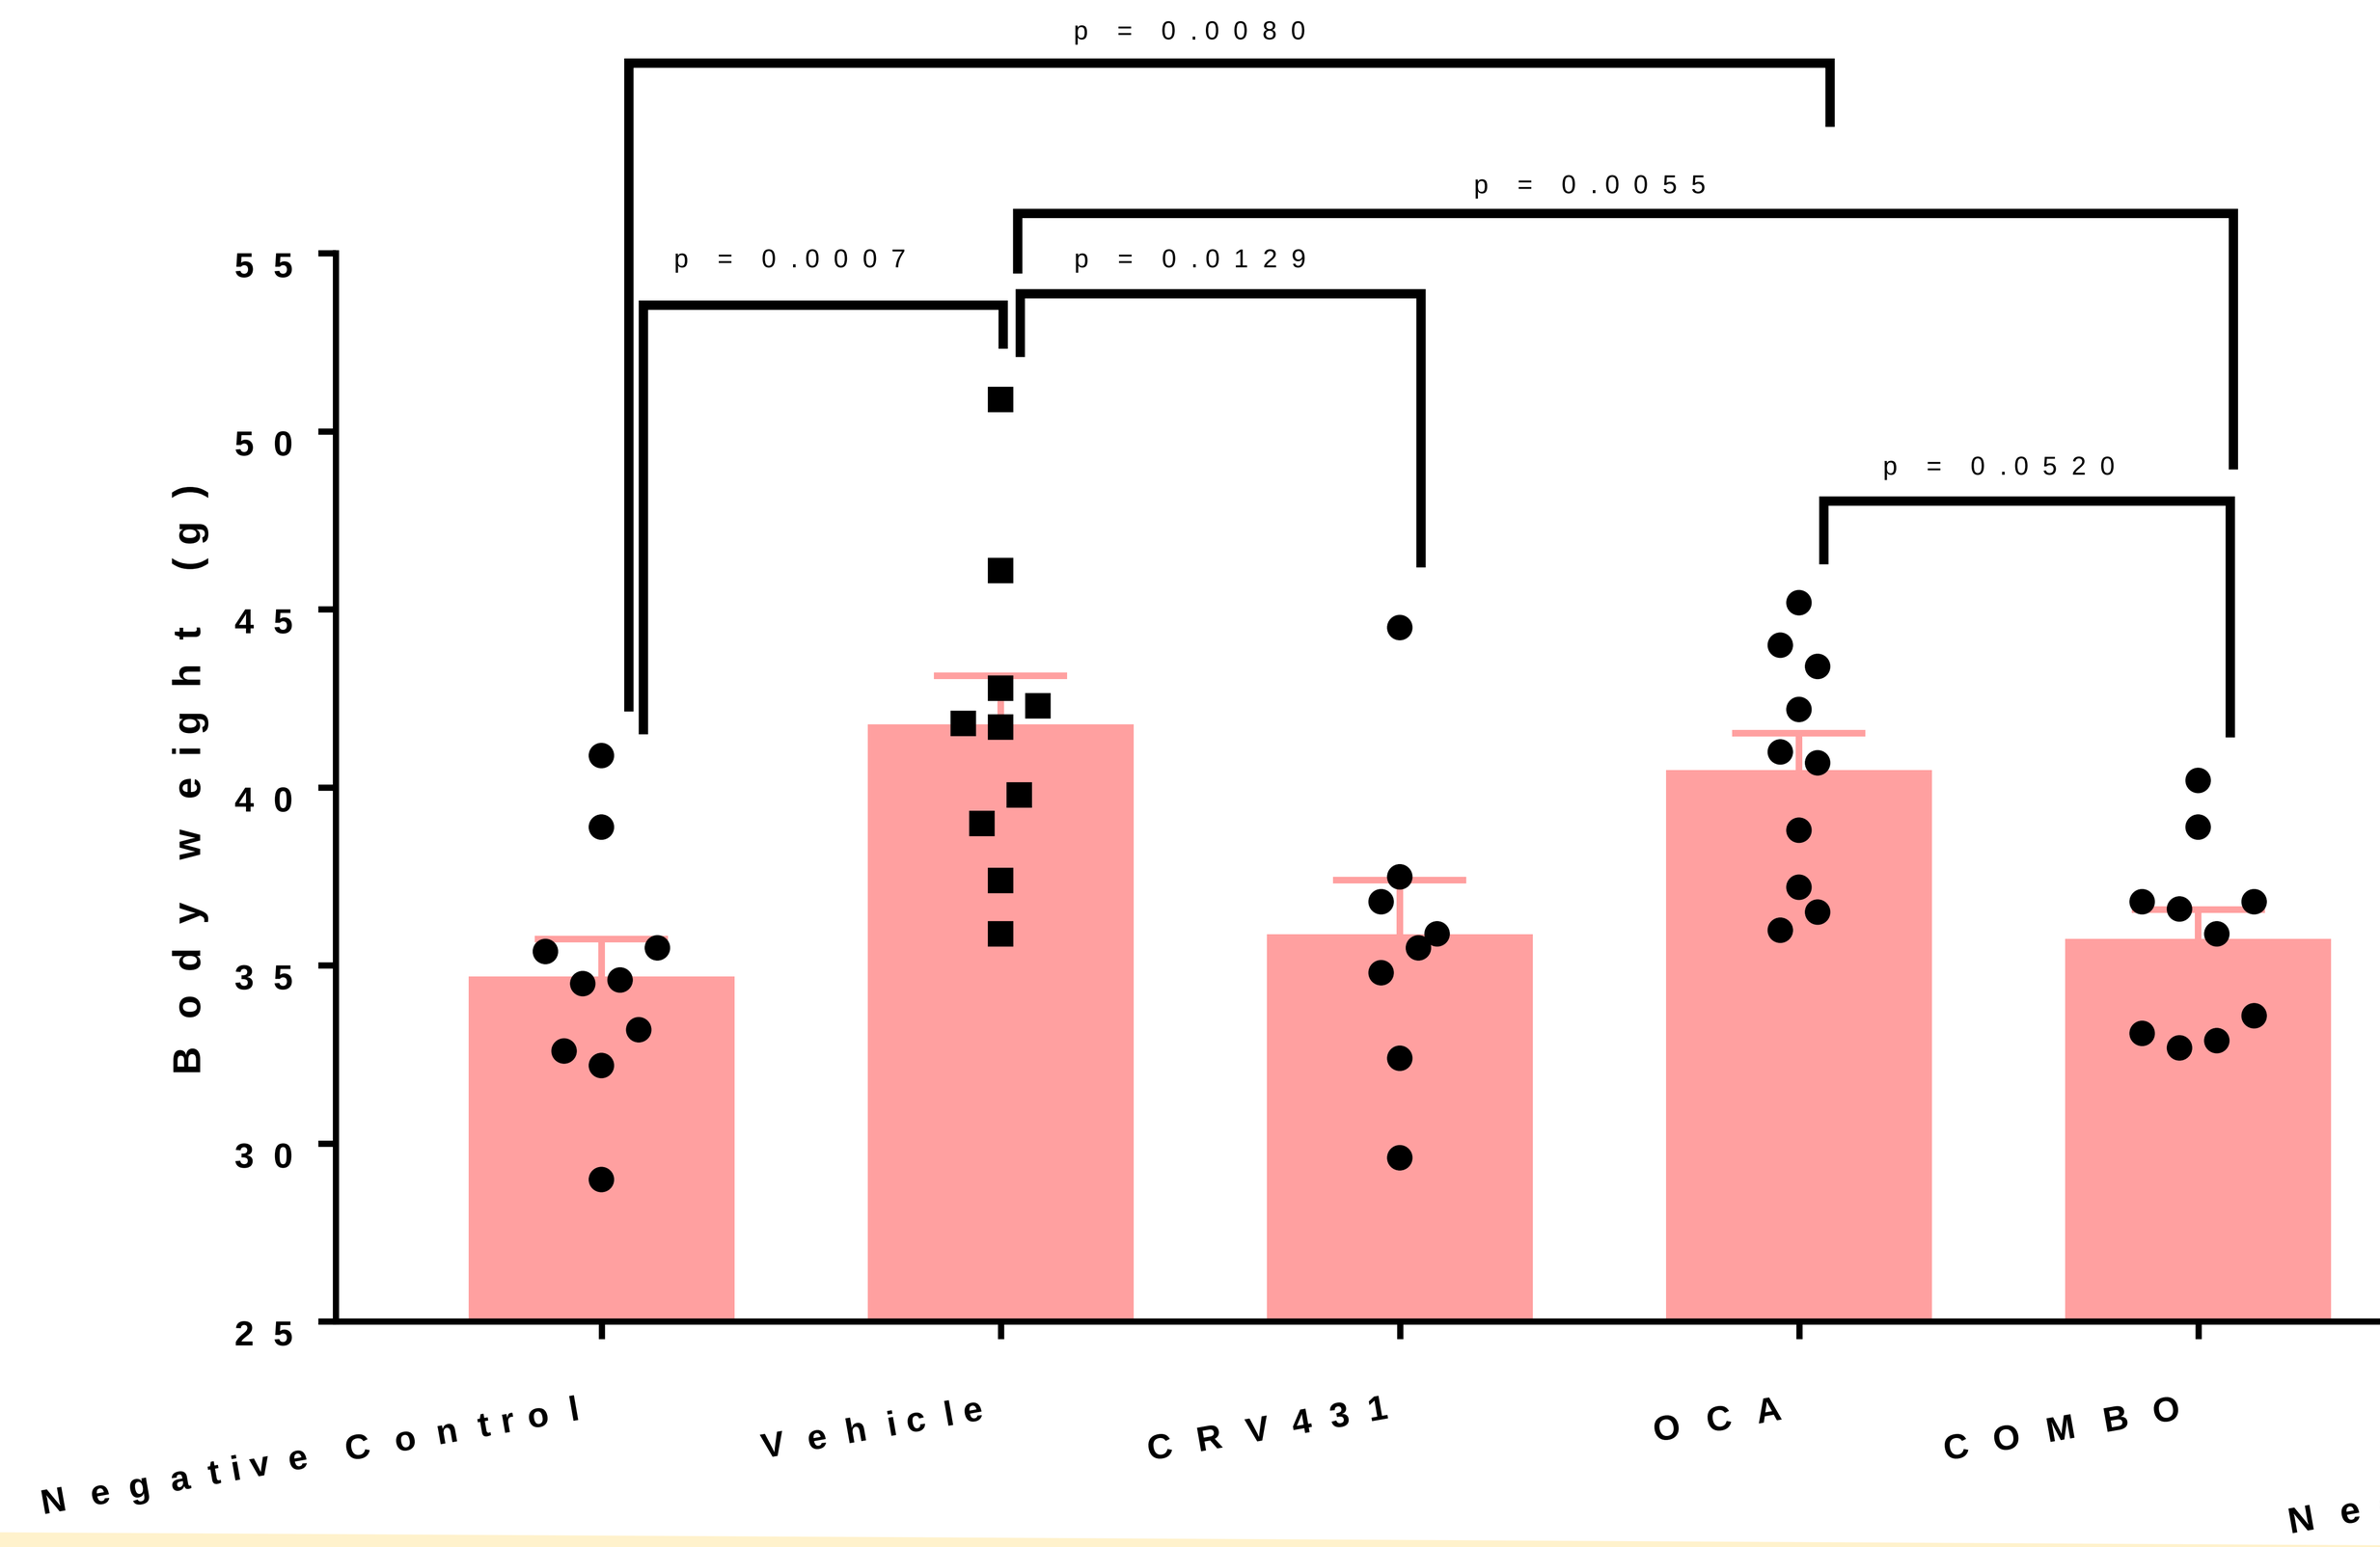
Body weight (g)
25
30
35
40
45
50
55
p = 0.0080
p = 0.0055
p = 0.0007
p = 0.0129
p = 0.0520
Negative Control
Vehicle
CRV431
OCA
COMBO

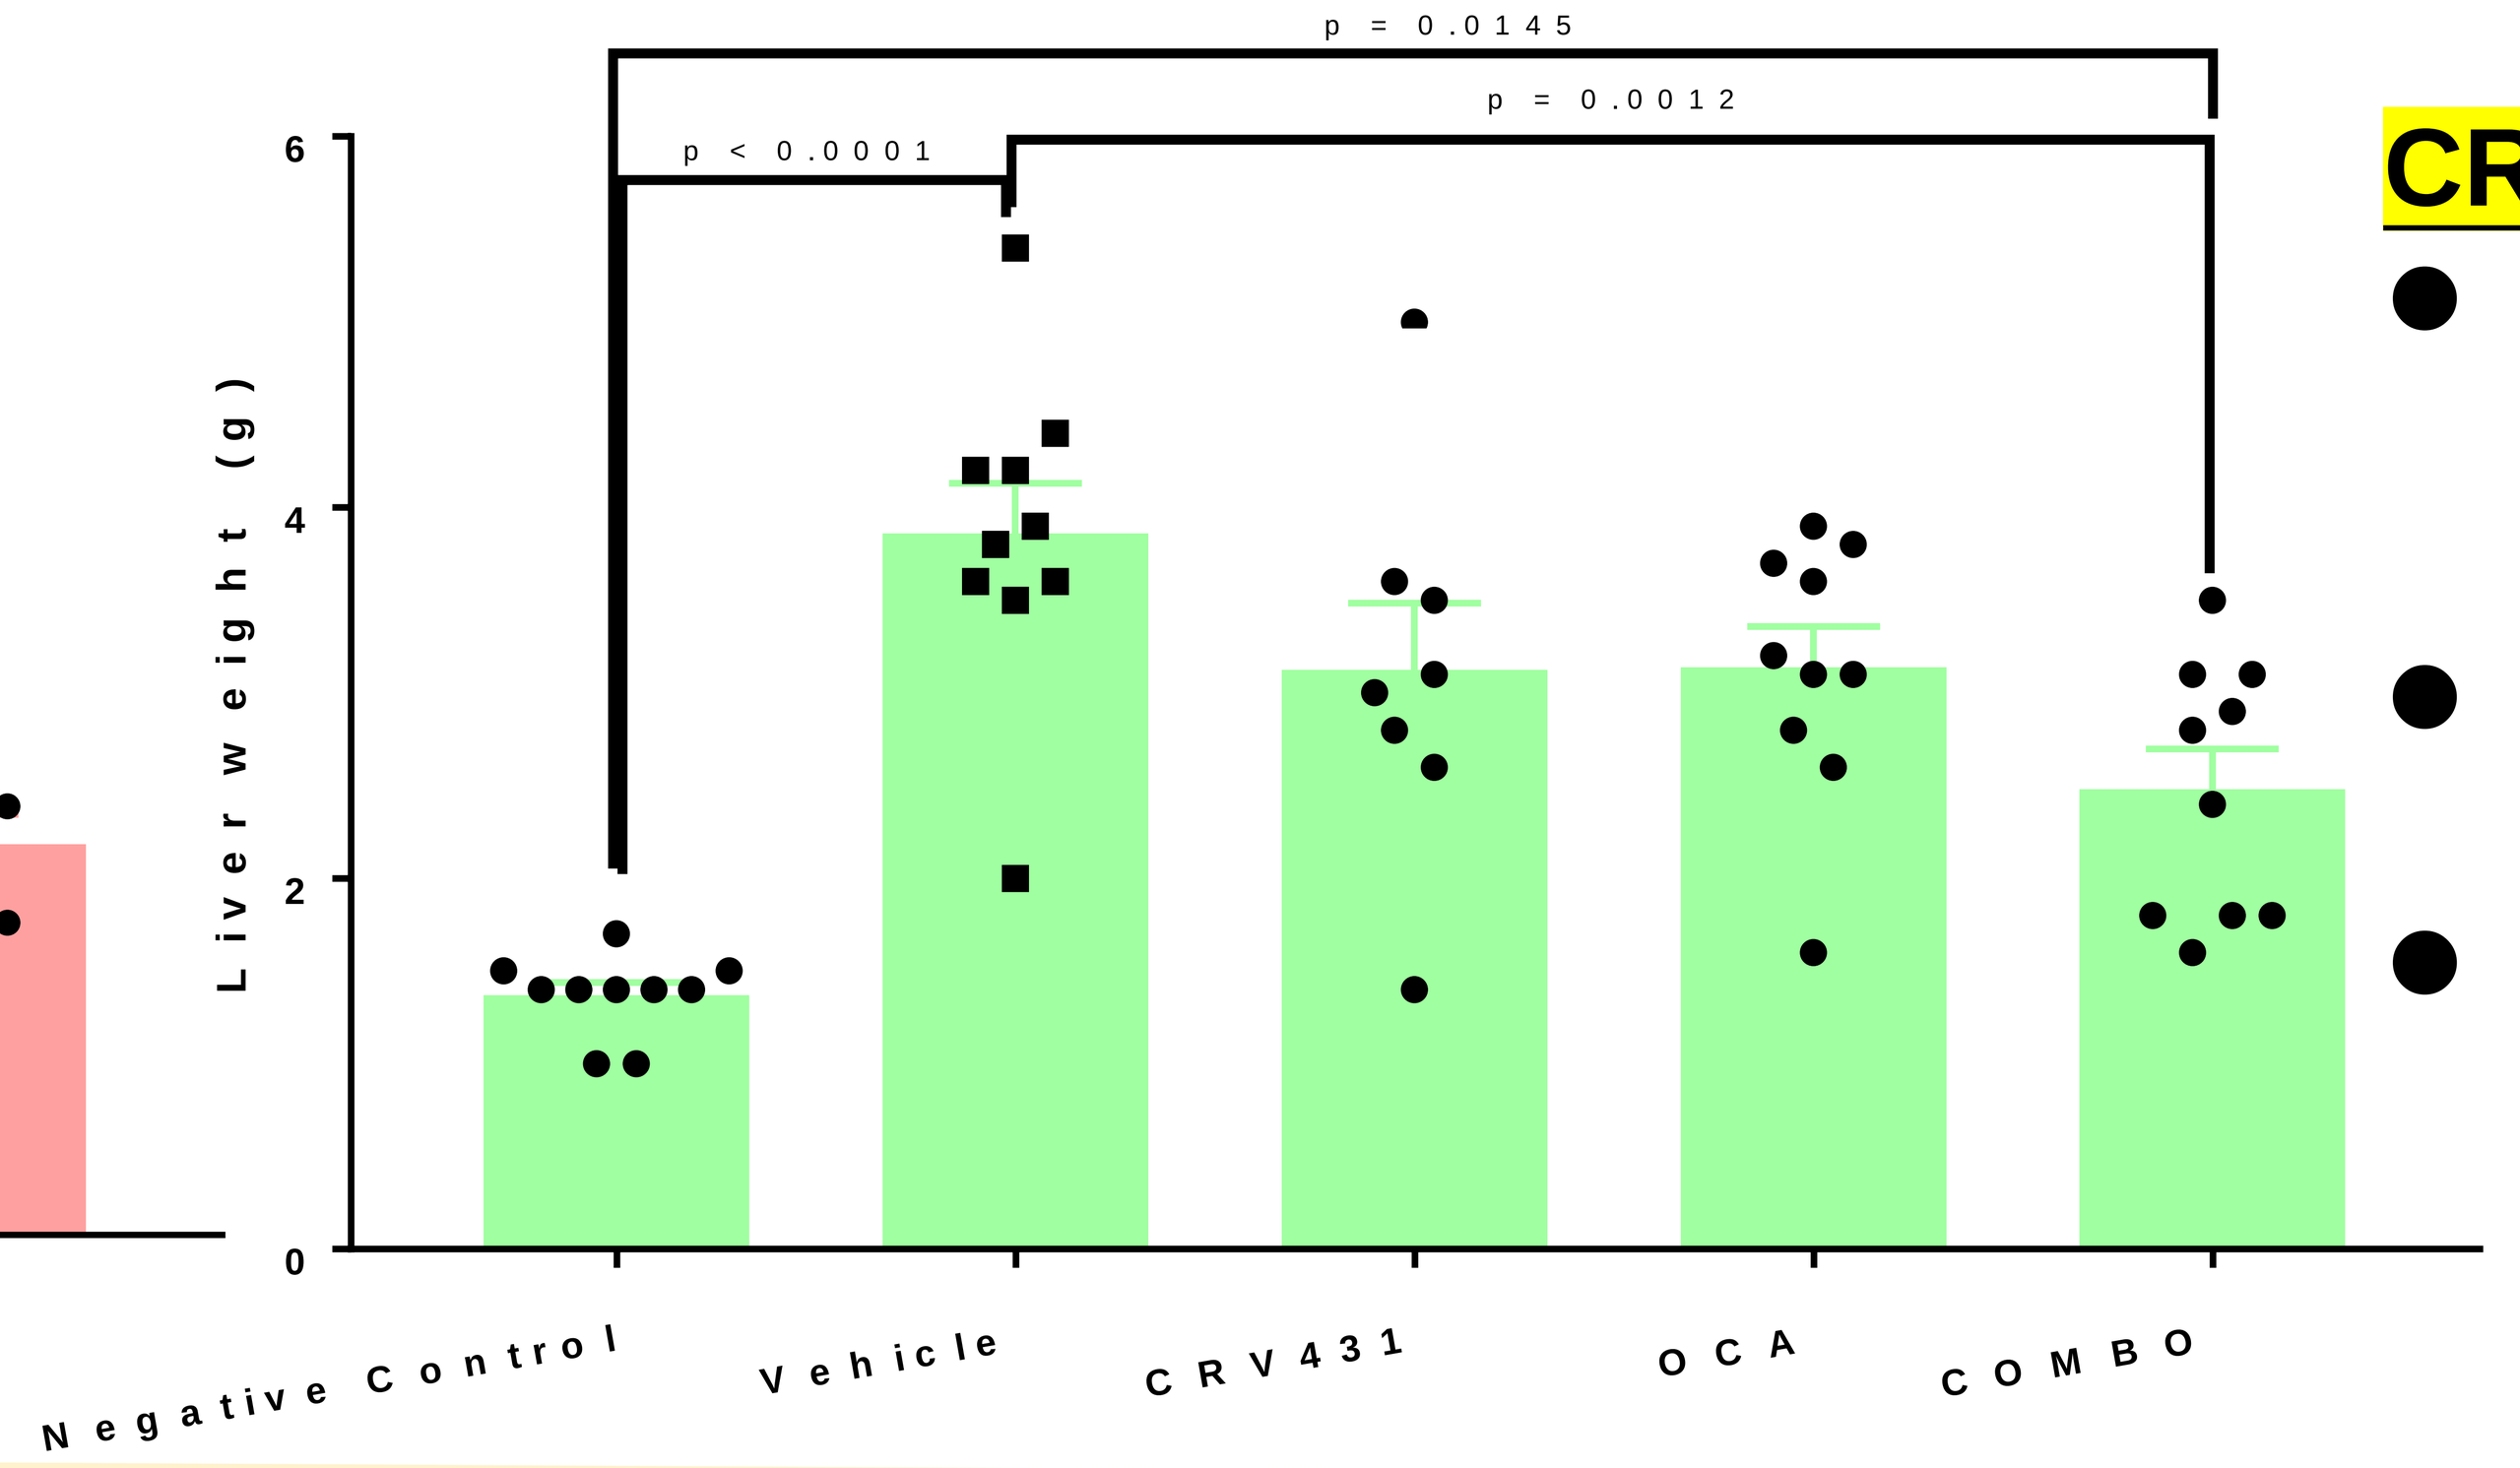
Liver weight (g)
0
2
4
6
p = 0.0145
p = 0.0012
p < 0.0001
Negative Control
Vehicle
CRV431
OCA
COMBO

CRV431 reduces body weight of mice

- CRV431 alone and COMBO lowered body weight compared to vehicle
- CRV431 or OCA alone did not decrease liver weight
- Decreased liver weight was seen in COMBO-treated mice

CONCLUSIONS

CRV431 is a potent agent against diet- and chemical-driven fibrosis. This result is consistent with our previous findings and suggests that cyclophilin inhibition can protect the liver from induced injuries.

This project was funded by the National Institute of Allergy and Infectious Diseases of the National Institutes of Health under award number R01AI125365 (Gallay). Kuo is supported by the Ruth L. Kirschstein Institutional National Research Service Award (T32AI007244). Kuo, Cabrera, and Gallay declare no competing financial interests. Mayo, Ure, and Foster are employees of Hepion Pharmaceuticals.